RAPID ROBUST MYCOPLASMA DETECTION SYSTEM FOR RELEASE TESTING OF CELL AND GENE THERAPY PRODUCTS CONTAINING MAMMALIAN CELLS IN 1-HOUR

Sandrine Sprugnoli[1], Doriane Piazza[1], Sophie Valery[1], Caroline Kassim Houssenaly[1], Ambili Menon[2]
[1]Pharma Quality Control, bioMérieux SA, Craponne, Rhône-Alpes, France • [2]Pharma Scientific Affairs, bioMérieux Inc, Lombard, IL, United States

PDA Pharmaceutical Microbiology Conference 2024 • 07 - 09 October 2024, Washington, DC
Scan the QR code to receive digital versions of our posters and whitepapers:

INTRODUCTION

Cell and gene therapies (C&GT) or Advanced Therapeutic Medicinal Products (ATMP) are innovative medicine developed to treat cancers, rare diseases, autoimmune disorders, and injuries. As these therapies utilize living cells, they produce a product with a short shelf life. While microbiological examination of cell-based products is critical to ensure patient safety, challenges exist to enable testing and obtaining rapid results prior to patient infusion.

One of these microbiological examinations is the mycoplasma testing of cellular therapy products which is a regulatory requirement for release. The compendial microbiological assay, currently recommended by the United States Pharmacopeia (USP), European Pharmacopeia (EP), Japanese Pharmacopeia (JP) and the US FDA, for mycoplasma testing of biologics, involves the culture of viable mycoplasmas in broth, agar plates and indicator cell cultures. This compendial culture testing utilizes complex media and requires ≥ 28 days to generate test results. Waiting 28 days or longer to release product isn't a feasible option for most autologous cell and gene therapy processes where patients are waiting to be infused and time is critical to effective treatment. **(1-3)**

Conventional nucleic acid testing (NAT) methods offer a faster alternative to compendial methods with results often within a single work shift; however, they remain highly manual, require specialized laboratories, elevated skill levels and significant training to execute the test and interpret results. Because of these complexities, mycoplasma testing is often outsourced to third-party laboratories, resulting in additional costs.

BIOFIRE® Mycoplasma: BioMérieux provides an innovative test for mycoplasma detection in cell and gene therapy products. The BIOFIRE® Mycoplasma test can detect the presence of >130 species of mycoplasma. The system provides sample to answer in ~1 hour with little technical training required. This provides an easy-to-learn, easy-to-use option to bring mycoplasma testing in-house, saving time and outsourcing costs.

Table 1: Comparison of TTR for various Mycoplasma testing methods.

Testing Method	Regulation	TTR	Expertise	Contamination risk	Reagent storage	Testing Space	Sensitivity	Sample Volume
BIOFIRE®	EP 9.0 <2.6.7>	<1 Hour	Novice	Low	Room Temp	PCR lab not needed	≤10 CFU/mL*	~1.7 mL-10mL
Other PCR Methods	USP 39 <63>	5-7 Hours	Expert	High	-20°C	PCR Lab		
Traditional Methods	JP 17 <G3>	6-28 Days	Expert	High	40°C	Specialized Lab		

PURPOSE

Introduce C&GT product specific protocols that allows the inclusion of mammalian cells thus aligning with the upcoming EP guideline **(4)** whilst providing the required level of detection (≤ 10 CFU/mL) as a Mycoplasma release test.

MATERIALS AND METHOD

BIOFIRE® MYCOPLASMA FILMARRAY® 2.0 INDUSTRY SYSTEM

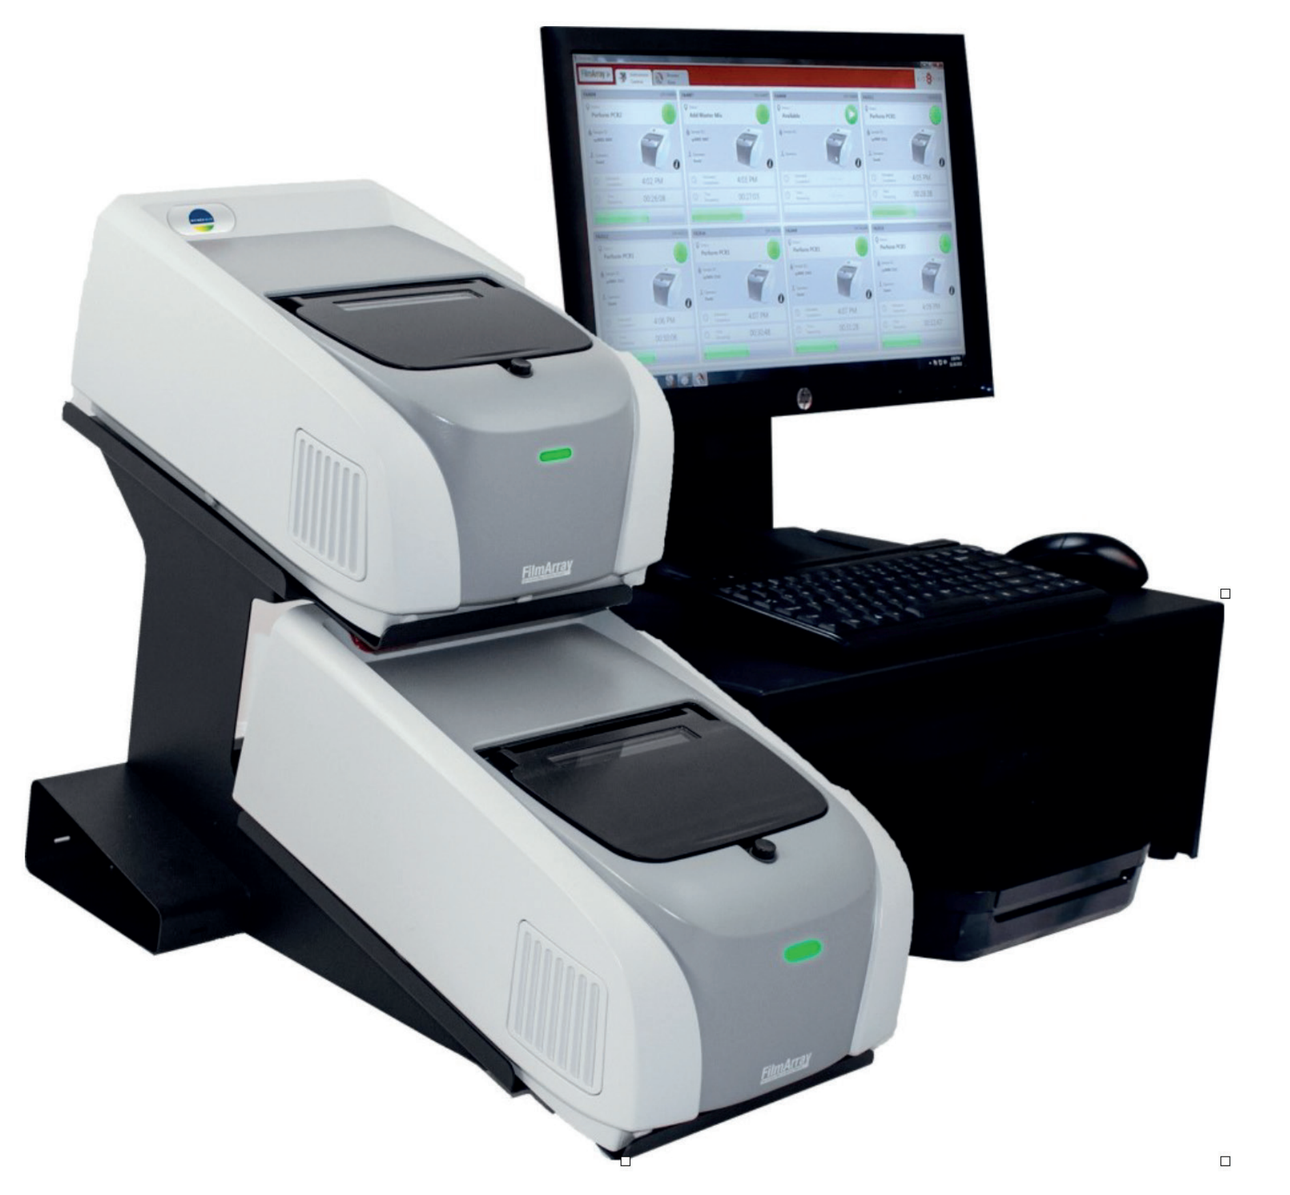

Figure 1: FILMARRAY 2.0 Industry instrument performs the extraction, amplification and detection (25.4 x 39.3 x 16.5 cm/10 x 15.5 x 6.5 in WxDxH). The system comes standard with 2 instruments; up to 8 instruments can be connected to a single PC.

The BIOFIRE system utilizes the FILMARRAY 2.0 Industry instrument and next generation PCR testing in a closed pouch to detect the presence of mycoplasma (Figure 1). The disposable BIOFIRE Mycoplasma pouch contains all the necessary reagents for automated PCR and Analyte detection to isolate, amplify and detect over 130 different mycoplasma species. Several controls are integrated into the pouch to ensure the quality of the results including a total process control, reverse transcription control, and PCR I and II controls (Figures 2A & 2B). The instrument & software process the pouch with results in less than an hour. The FILMARRAY 2.0 Industry software (21 CFR Part 11 compliance ready) performs all the complex Meta-analysis and provides presence/absence results as either "Mycoplasma Detected" or "Mycoplasma Not Detected".

2A

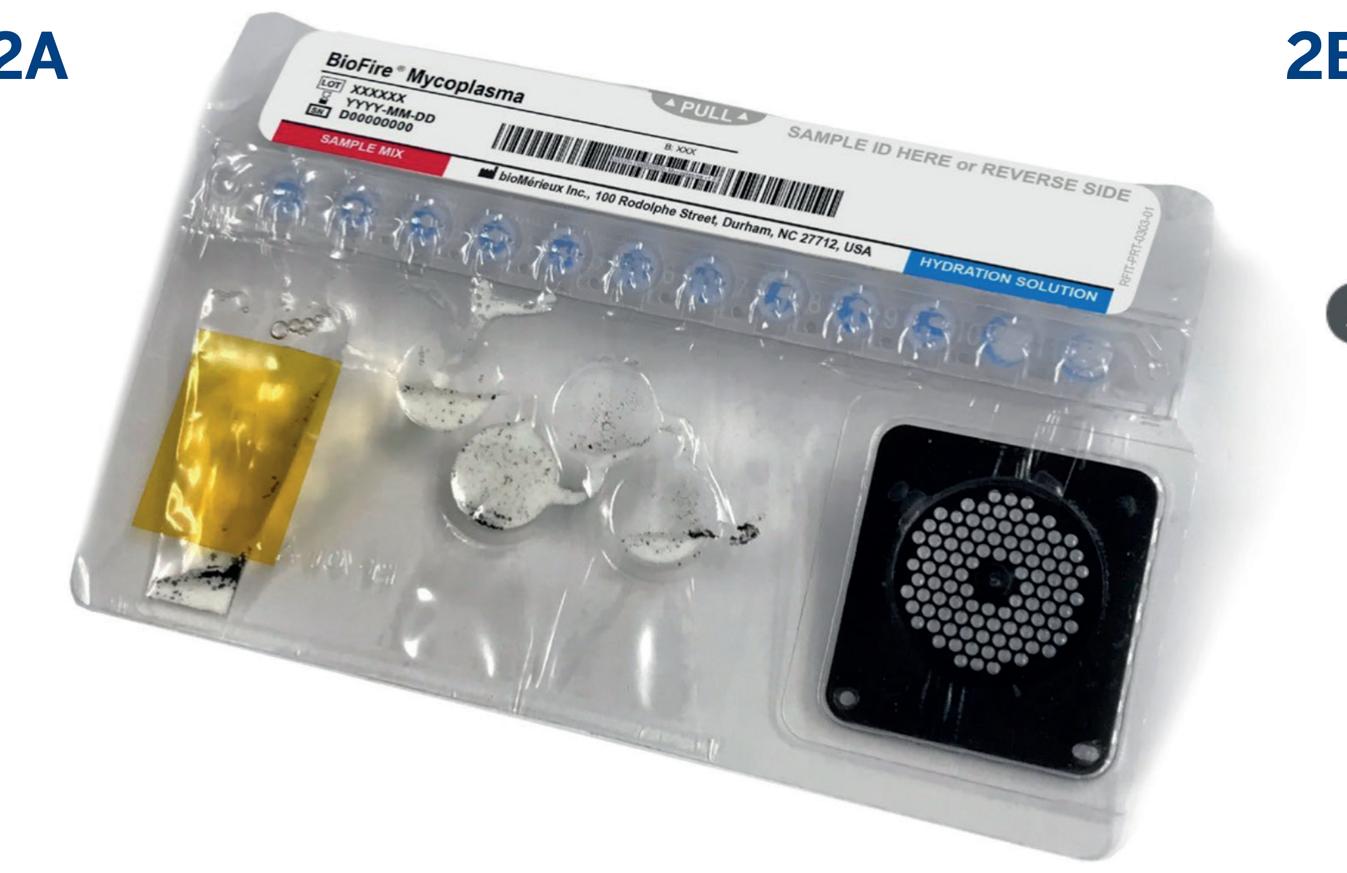

2B

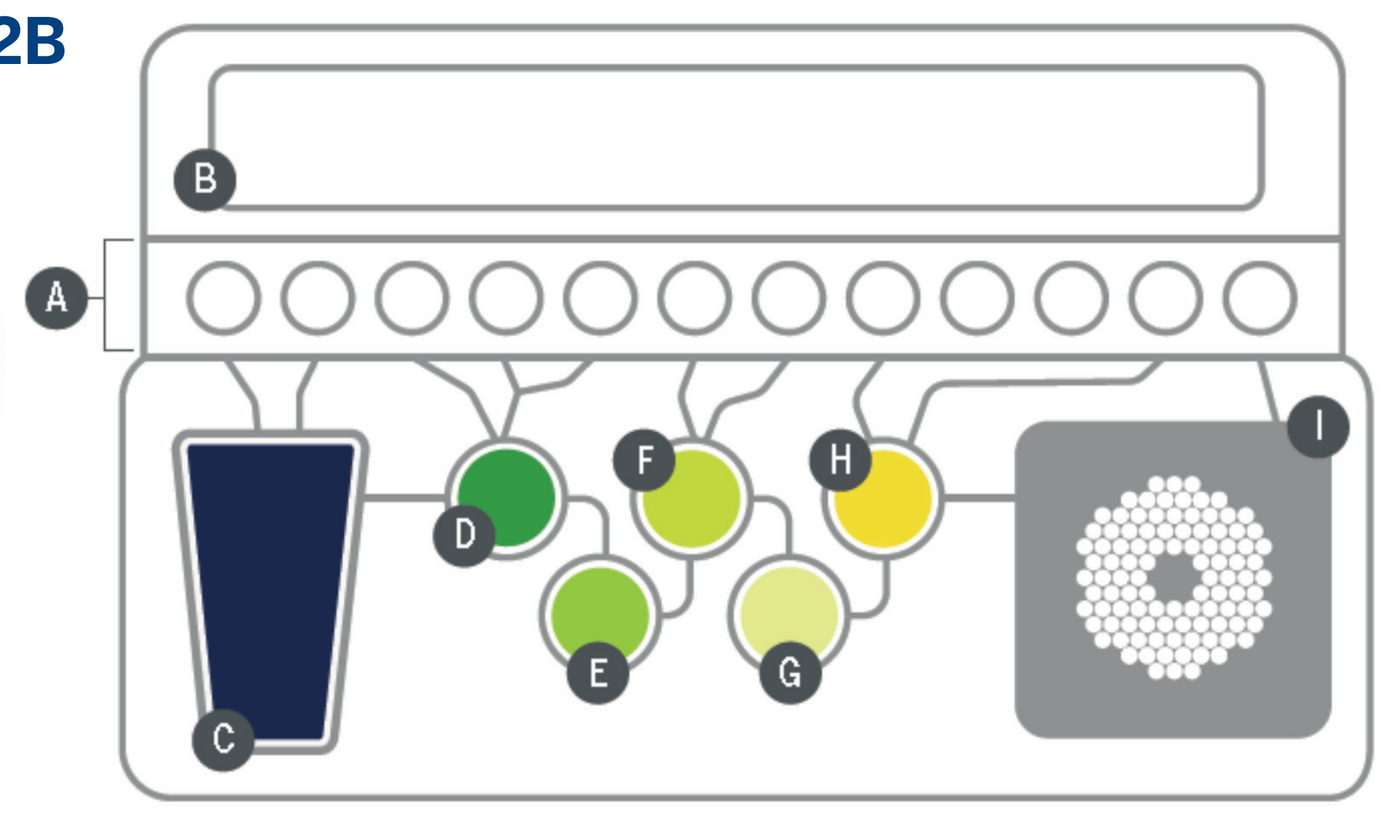
A
B
C
D
E
F
G
H
I

*Figure 2: **2A.** BIOFIRE Mycoplasma pouch. **2B.** Pouch diagram (A). Fitment with freeze-dried reagents (B). Plungers-deliver reagents to blisters (C). Sample lysis and bead collection (D). Wash (E). Magnetic bead collection blister (F). Elution (G). Multiplex Outer PCR blister (H). Dilution blister (I). Inner Nested PCR array.*

Sample Protocols for C&GT Products

Different Pharmacopeias around the world, including the USP and the upcoming EP release indicate testing C&GT products in the presence of cellular matrix. To align with these regulatory guidelines, two sample preparation protocols have been developed: the 10 mL single centrifugation protocol and the low volume (~1.7 mL) protocol for manufacturers with limited test article availability (Figure 3). Both protocols allow the inclusion of mammalian cells and achieve appropriate sensitivity at concentrations needed as a release test of a wide range of C&GT products.

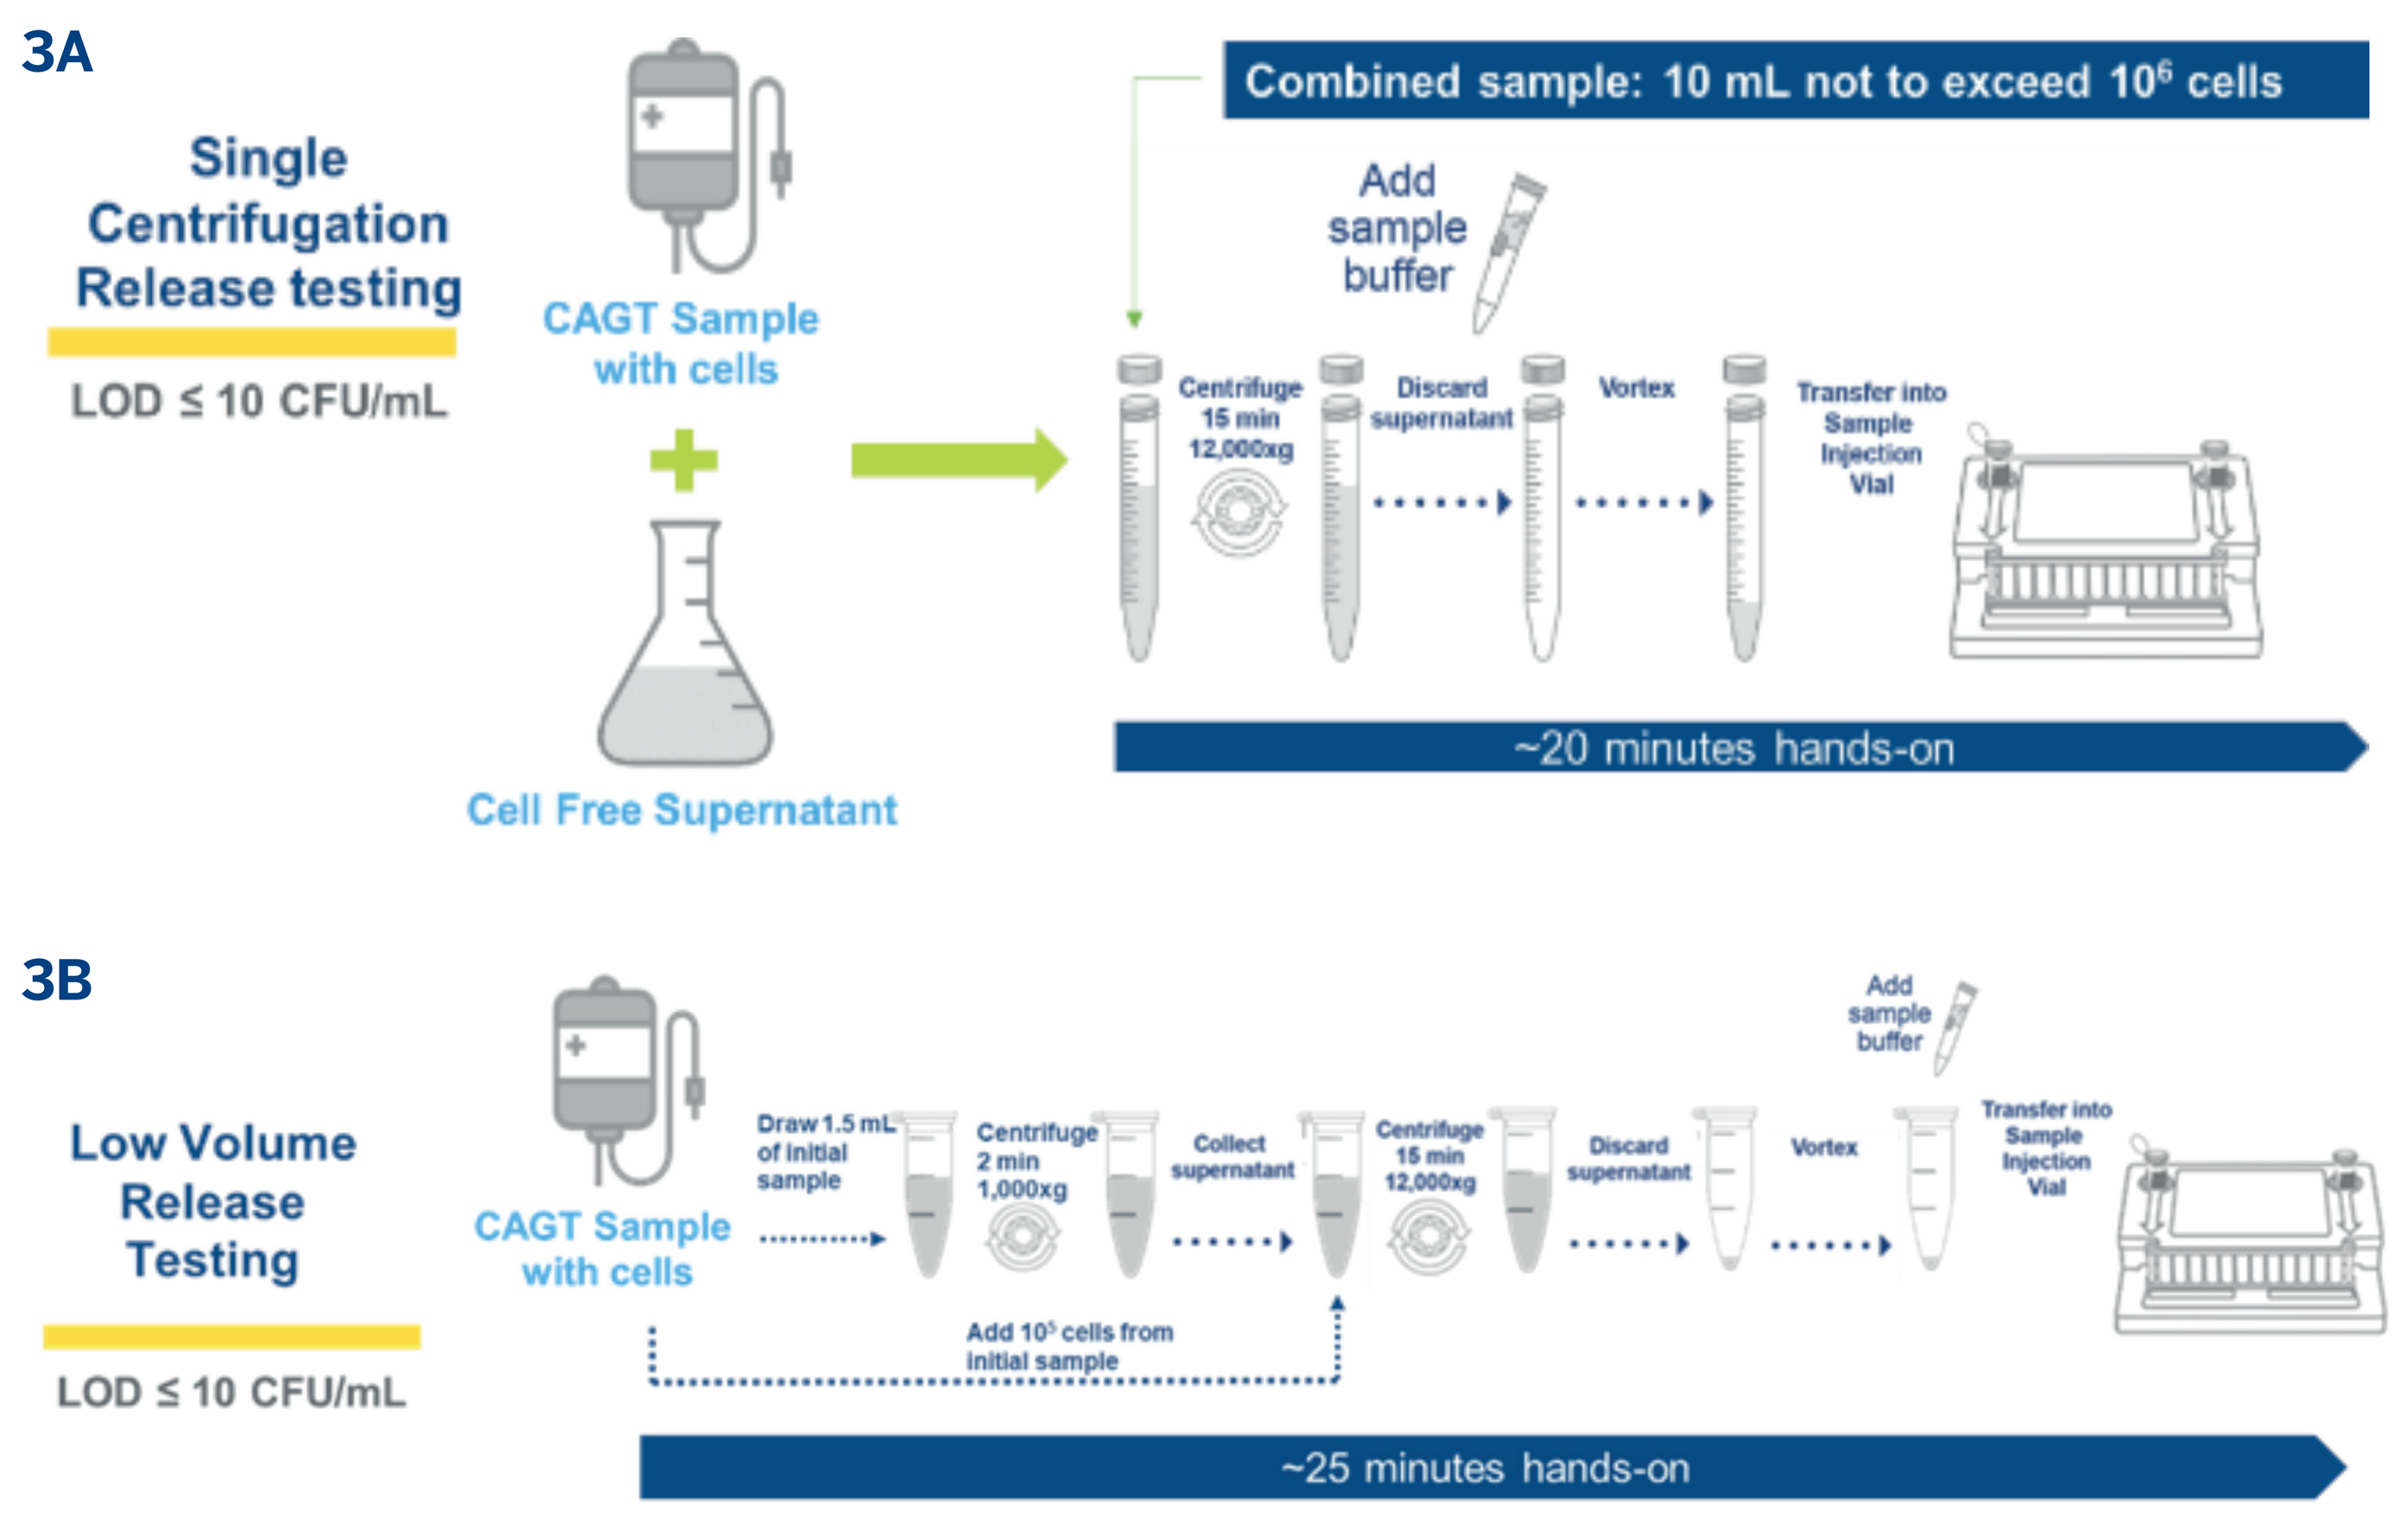
3A
Single Centrifugation Release testing
LOD ≤ 10 CFU/mL
CAGT Sample with cells
Cell Free Supernatant
Combined sample: 10 mL not to exceed 10⁶ cells
Add sample buffer
Centrifuge 15 min 12,000xg
Discard supernatant
Vortex
Transfer into Sample Injection Vial
~20 minutes hands-on
3B
Low Volume Release Testing
LOD ≤ 10 CFU/mL
CAGT Sample with cells
Draw 1.5 mL of initial sample
Centrifuge 2 min 1,000xg
Collect supernatant
Centrifuge 15 min 12,000xg
Discard supernatant
Vortex
Add sample buffer
Transfer into Sample Injection Vial
Add 10⁵ cells from initial sample
~25 minutes hands-on

*Figure 3: BIOFIRE FILMARRAY 2.0 Industry system release protocols for Mycoplasma testing with cells addition. **3A.** 10 mL Single centrifugation; **3B.** Low volume pre-processing for 1.7 mL samples – Double centrifugation.*

Sample preparation

10 mL of Jurkat cells at 10^5 cells/mL were spiked with live mycoplasma at 10 CFU/mL and processed following the 10 mL single centrifugation protocol. The low volume (~1.7 mL) sample preparation protocol has been designed for situations where product availability is limited, and post-expansion broth is not available. This protocol comprises 2 steps: a first step to normalize the cell density and a second step to concentrate the sample prior to loading into the pouch. The performance of this low volume protocol was assessed on Jurkat cells at 10^7cells/mL. Both studies involved 24 replicates for each mycoplasma strain tested with multiple independent dilution events, days, operators & instruments.

***Table (2A):** Mycoplasma detection in 10^5 Jurkat cells/mL using the 10 mL single centrifugation protocol at a target 10 CFU/mL.*

Protocol tested	Mycoplasma strain (ATCC-TTR)	Number of positive replicates	Detection rate %
10 mL Single Centrifugation Protocol	*Acholeplasma laidlawii*	24/24	100
	Mycoplasma arginini	23/24	95
	Mycoplasma fermentans	24/24	100
	Mycoplasma gallisepticum	24/24	100
	Mycoplasma hominis	23/24	95
	Mycoplasma hyorhinis	23/24	95
	Mycoplasma pneumoniae	24/24	100
	Mycoplasma orale	24/24	100
	Mycoplasma salivarium	24/24	100
	Mycoplasma synoviae	24/24	100

***Table (2B):** Mycoplasma detection in 10^5 Jurkat cells/mL using the 10 mL single centrifugation protocol at a target 10 CFU/mL.*

Protocol tested	Mycoplasma Strains (ATCC-TTR or *Mycosafe)	Number of positive replicates	Detection rate %
Low Volume (~1.7 mL) protocol	*Acholeplasma laidlawii*	23/24	95
	Mycoplasma arginini	24/24	100
	Mycoplasma fermentans	24/24	100
	Mycoplasma hyorhinis BTS7	24/24	100
	*Mycoplasma hyorhinis Alpha**	23/24	95
	Mycoplasma orale	23/24	95
	Mycoplasma salivarium	24/24	100
	*Mycoplasma pneumoniae**	24/24	100

FEASIBILITY STUDIES

These protocols were also evaluated on C&GT customer samples with multiple mycoplasma strains. No false positive results were observed, and mycoplasma detection was achieved when spiked at LOD 10 CFU/mL for all the sample matrices **(Table 3).**

***Table 3:** Feasibility studies on C&GT matrices*

Sample type	Protocol tested	Sample compatibility without spiking (N=3)	Inoculation study	
			Organisms LOD: 10 CFU/mL	Mycoplasma detection in product (N=3 per strain)
Car-T cells (1)	*10 mL Single Centrifugation protocol*	Pass	*A. laidlawii, M. fermentans, M. gallisepticum, M. synoviae*	100%
Car-T cells (2)		Pass	*A. laidlawii*	100%
Car-T cells (3)		Pass	*M. hominis, M. arginini*	100%
Car-T cells (4)		Pass	*M. pneumoniae*	100%
Car-T cells (5)		Pass	*M. pneumoniae,M. orale, M. hyorhinis, M. fermentans*	100%
Mesenchymal cells		Pass	*M.orale*	100%
Dendritic cells		Pass	*M.orale*	100%
Multi TAA specific T-cells		Pass	*M.orale*	100%
Human Kidney cells		Pass	*A. laidlawii,*	100%
Car-T cells (5)	*Low volume (~1.7 mL) protocol*	Pass	*M. pneumoniae, M. orale, M. hyorhinis, M. fermentans*	100%

CONCLUSION

The results of these studies show that the BIOFIRE FILMARRAY 2.0 Industry system is well suited as a release test for cell and gene therapy products. The results showed high sensitivity using two sample preparation options; one for testing 10 mL of product and a low volume protocol that only requires 1.7 mL of test article. Both protocols showed detection of a comprehensive panel of pharmacopoeia mycoplasma strains providing reliable results in less than 1 hour.

In addition, compatibility across a wide range of commonly used cell types was shown giving the ability for the cell and gene therapy markets to perform fast and simple screening for mycoplasma contamination using the BIOFIRE Mycoplasma test.

REFERENCES:

1. United States Pharmacopeia. 2022. General chapter <63> Mycoplasma tests
2. European Pharmacopeia. 2008. Chapter 2.6.7. Mycoplasmas
3. Japanese pharmacopoeia 18th edition
4. European Pharmacopeia. 2022. Chapter 2.6.7. Mycoplasmas, draft
5. BIOFIRE Mycoplasma Panel Instructions for Use, REF 423306, Part Number DFA2-PRT-0051-05

biomerieux.com
PIONEERING DIAGNOSTICS