An Integrated Lifecycle Approach to Contamination Control Strategy (CCS) Using Quality Risk Management (QRM) Principles with a Case Study

Darshana Patel, Associate Director, Micro Quality & Sterility Assurance
Merck & Co., Inc., Rahway, NJ, USA
Contact email: darshana_patel@merck.com

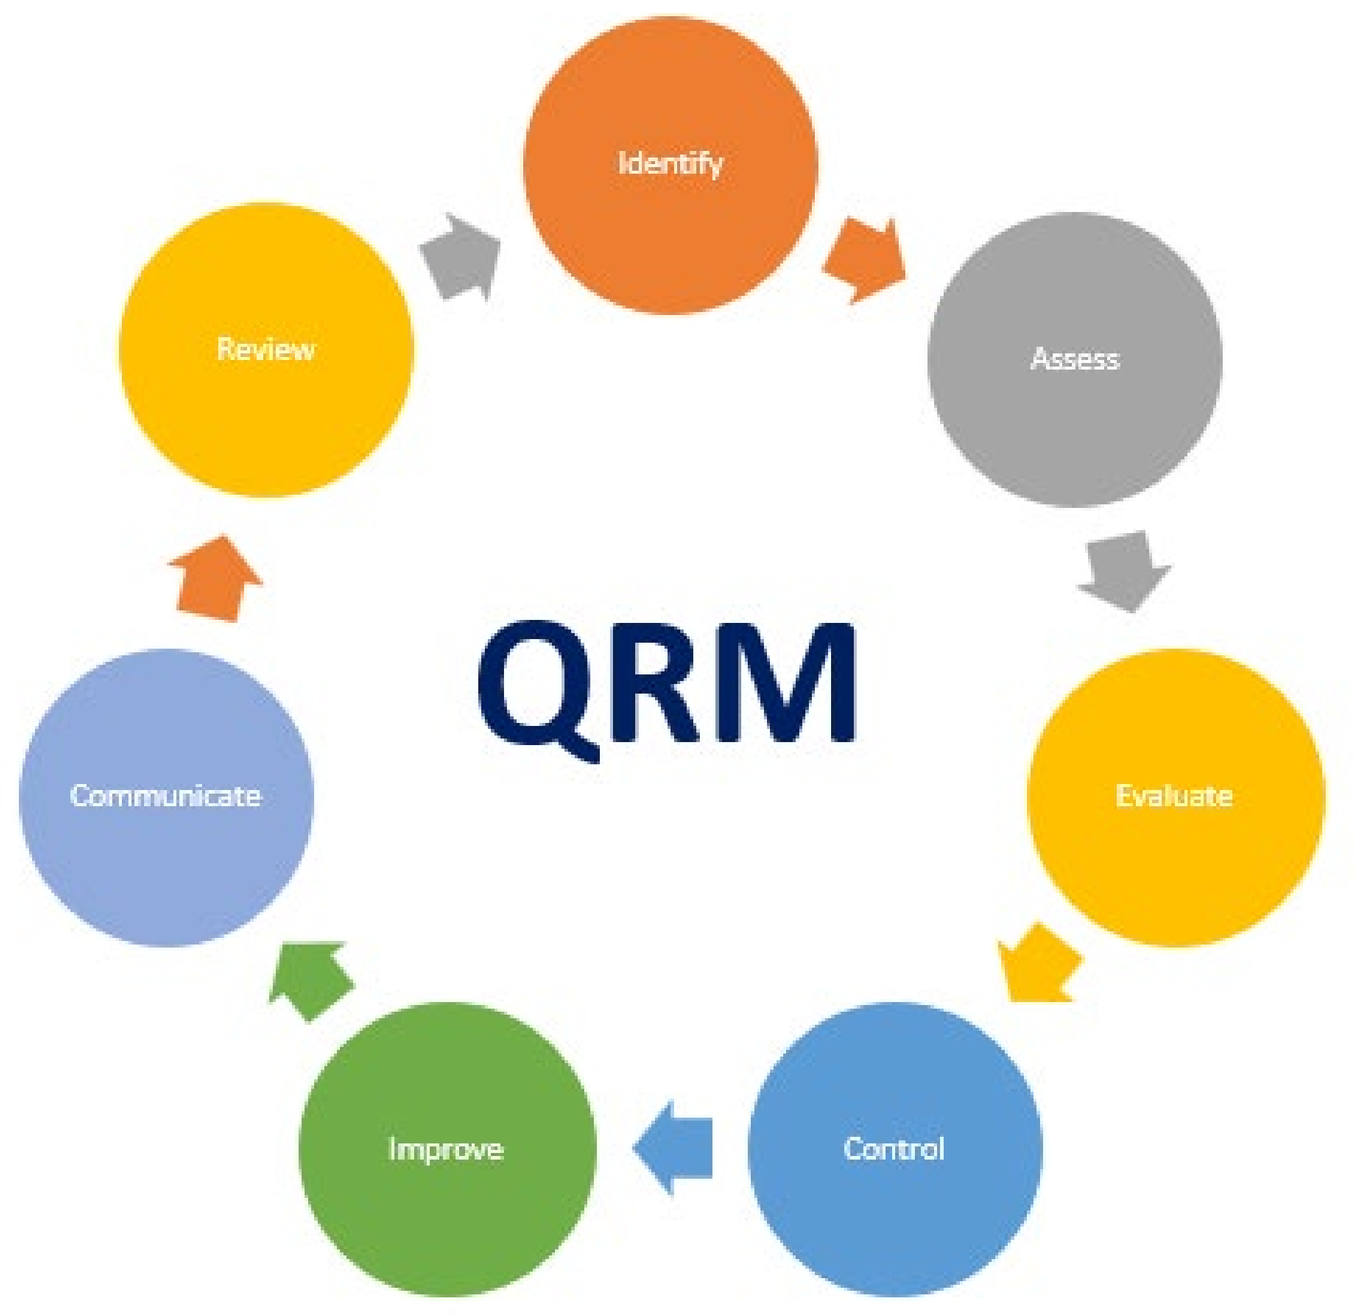
QRM
Identify
Assess
Evaluate
Control
Improve
Communicate
Review

Introduction

Contamination control is a critical aspect of ensuring that the manufacturing process in a pharmaceutical industry is designed to meet product quality, safety, and efficacy as well as ensuring compliance with regulatory expectations.

"A Contamination Control Strategy (CCS) should be implemented across the facility in order to define all critical control points and assess the effectiveness of all the controls (design, procedural, technical and organizational) and monitoring measures employed to manage risks to medicinal product quality and safety."[1]

Using a cohesive structure of QRM process and integration of the key components, can lead to development of a comprehensive Contamination Control Strategy (CCS).

CCS should consider all integral elements of sterile product manufacturing, aseptic preparation, including QRM principles and supporting risk assessments of contamination control and monitoring. It should describe all control measures required to prevent /reduce risk of microbiological, non-viable particulate, extraneous matter, product-product, and endotoxin/pyrogenic contamination.

A life cycle management strategy should be established for CCS and consideration should be given to design, development, manufacturing process controls, and ongoing continuous improvements of the product.

Quality Risk Management (QRM)

"A systematic process used to assess, control, communicate and review risks related to quality of a medicinal product throughout its lifecycle."[3]

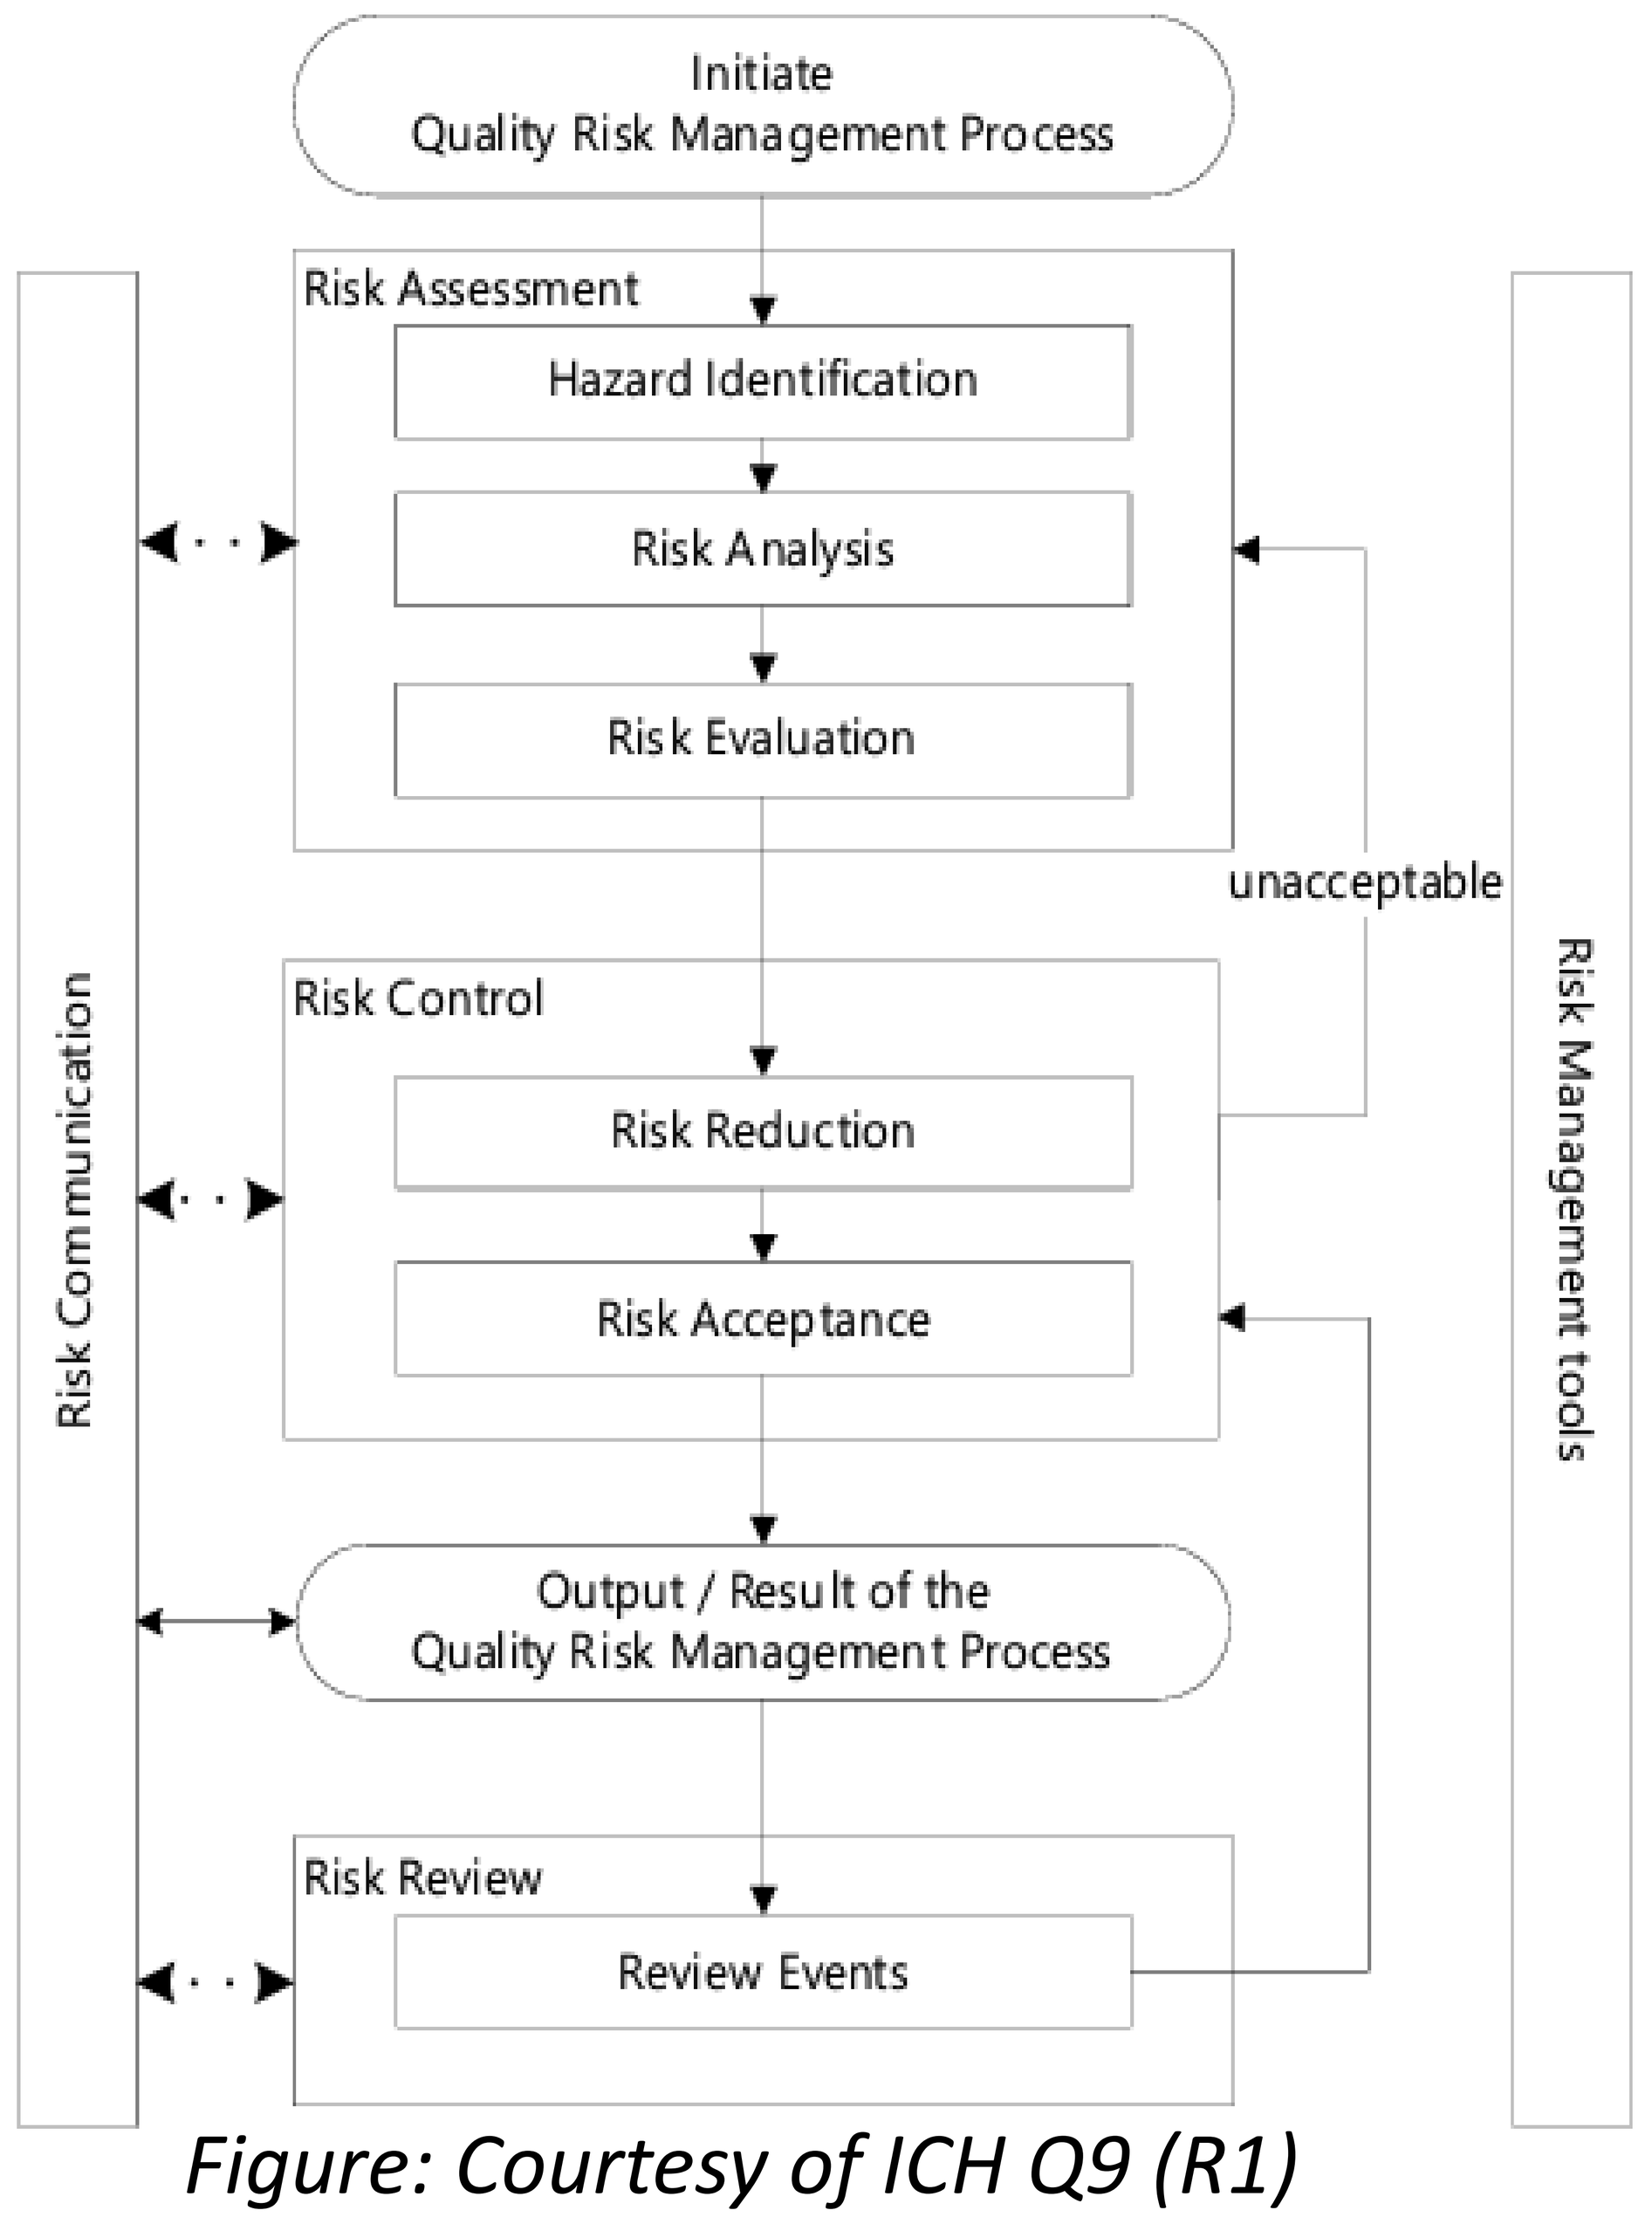
Initiate
Quality Risk Management Process
Risk Assessment
Hazard Identification
Risk Analysis
Risk Evaluation
unacceptable
Risk Control
Risk Reduction
Risk Acceptance
Output / Result of the
Quality Risk Management Process
Risk Review
Review Events
Risk Communication
Risk Management tools

Figure: Courtesy of ICH Q9 (R1)

QRM Process

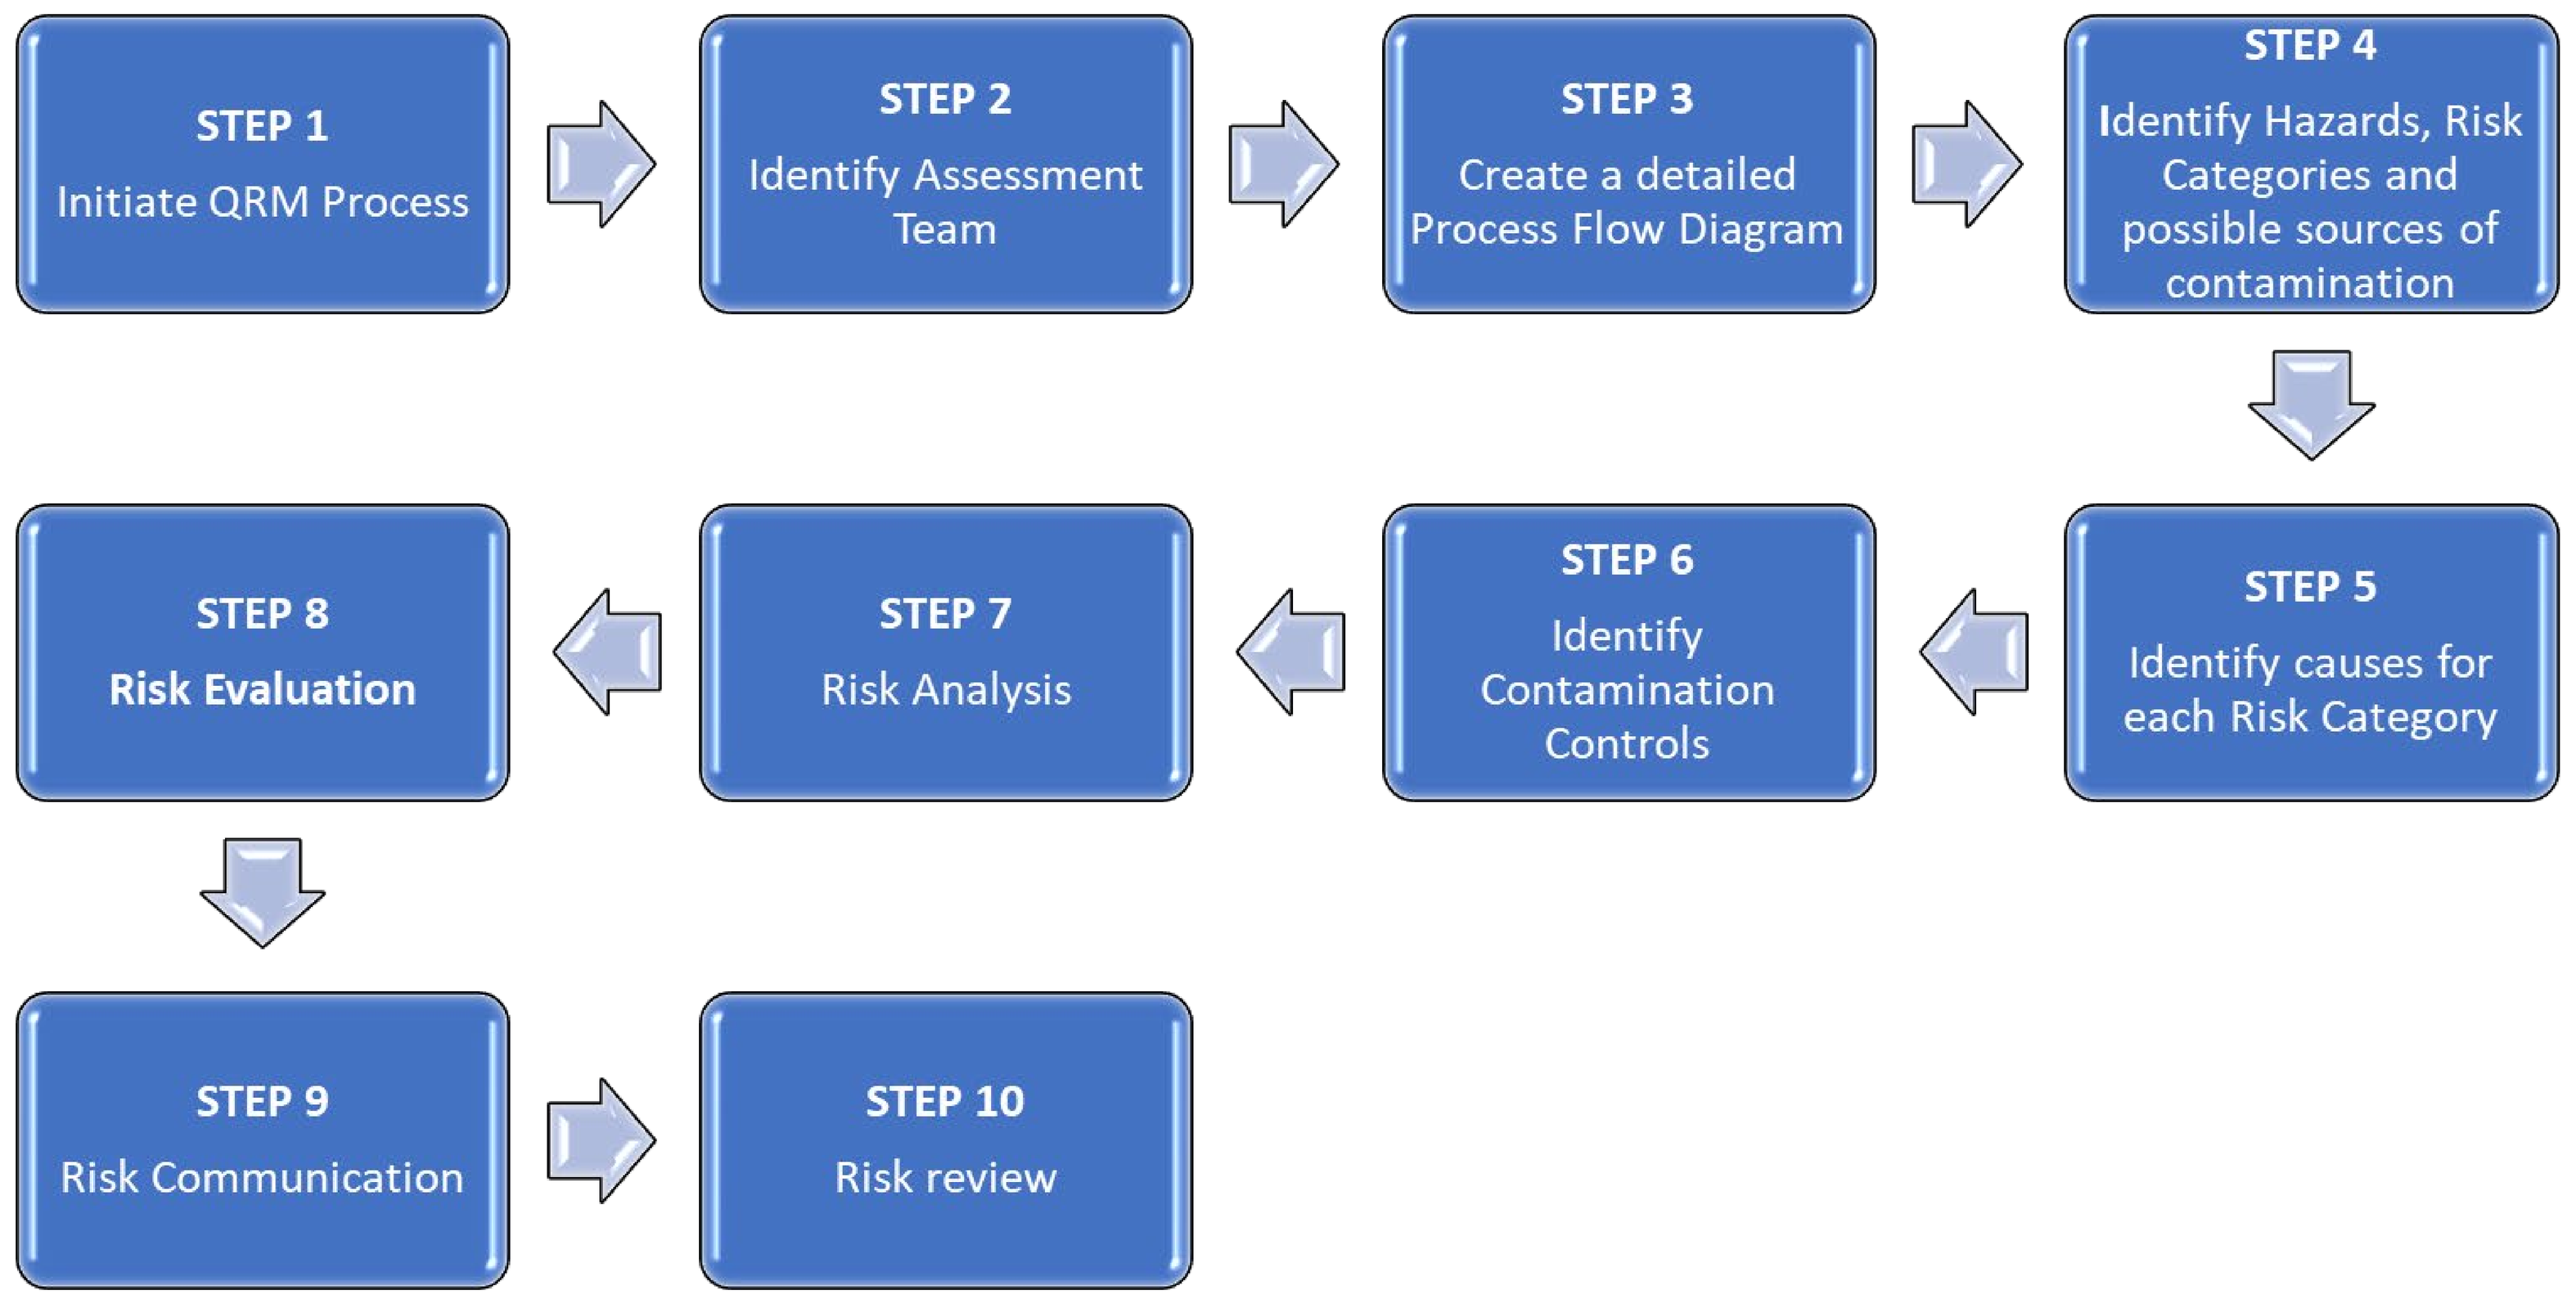
STEP 1
Initiate QRM Process
STEP 2
Identify Assessment Team
STEP 3
Create a detailed Process Flow Diagram
STEP 4
Identify Hazards, Risk Categories and possible sources of contamination
STEP 5
Identify causes for each Risk Category
STEP 6
Identify Contamination Controls
STEP 7
Risk Analysis
STEP 8
Risk Evaluation
STEP 9
Risk Communication
STEP 10
Risk review

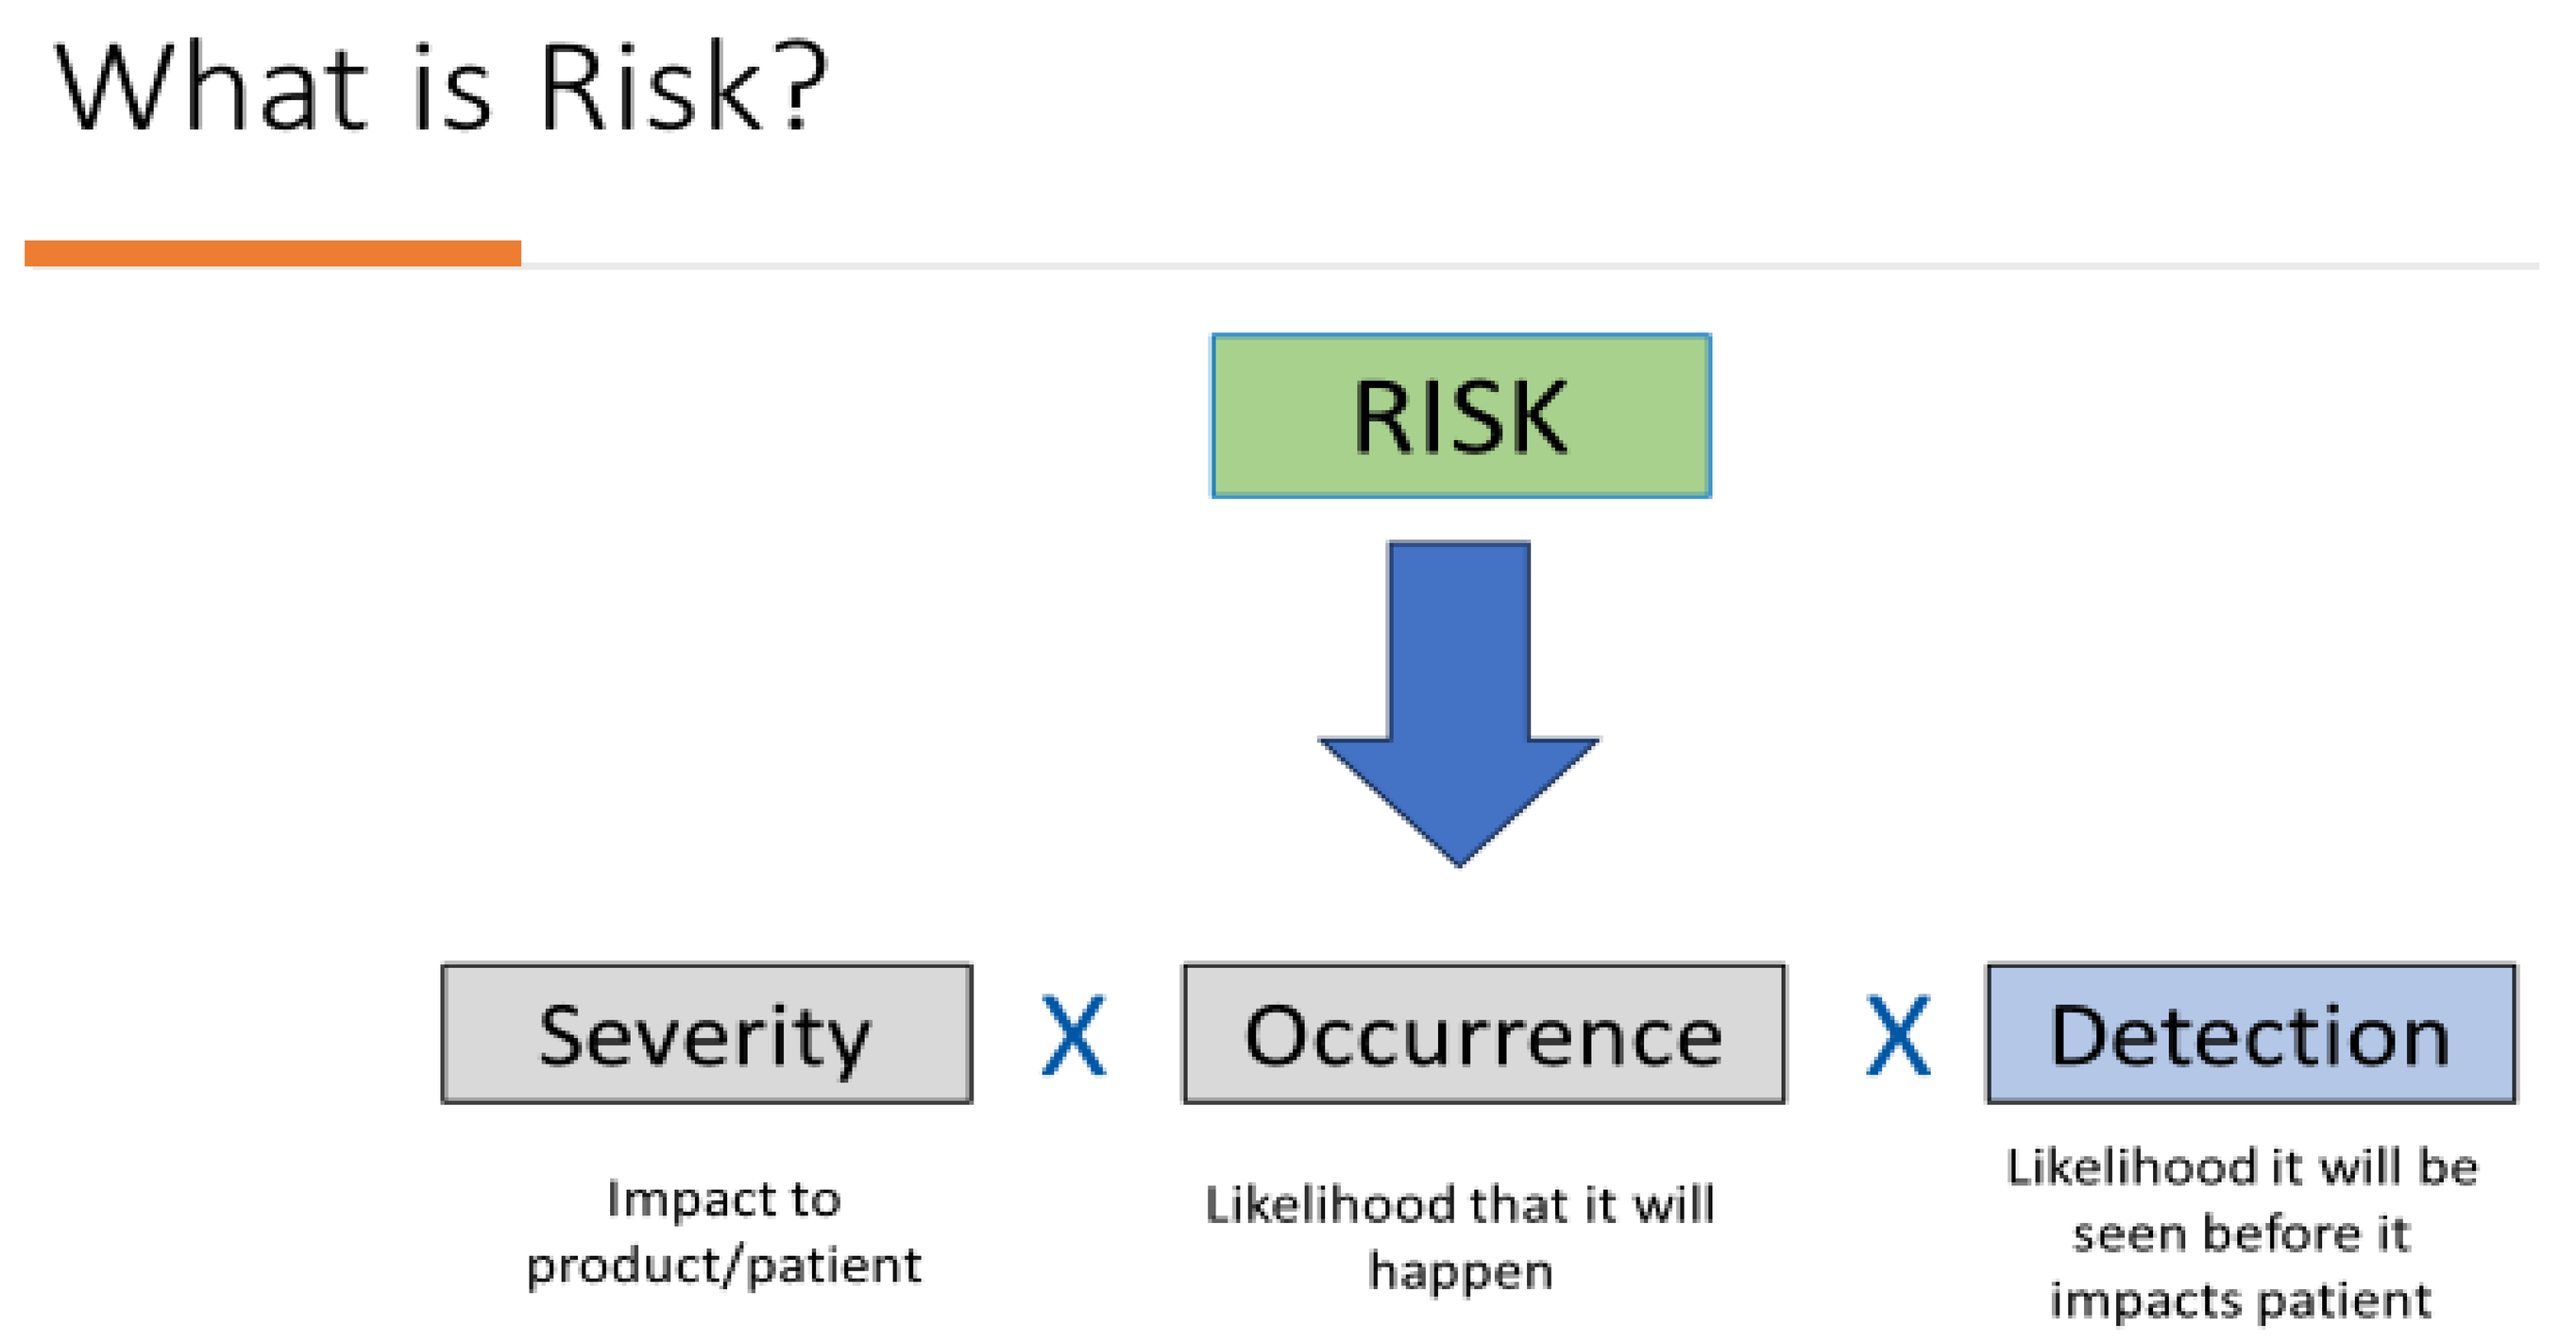
What is Risk?
RISK
Severity
X
Occurrence
X
Detection
Impact to product/patient
Likelihood that it will happen
Likelihood it will be seen before it impacts patient

Elements of CCS

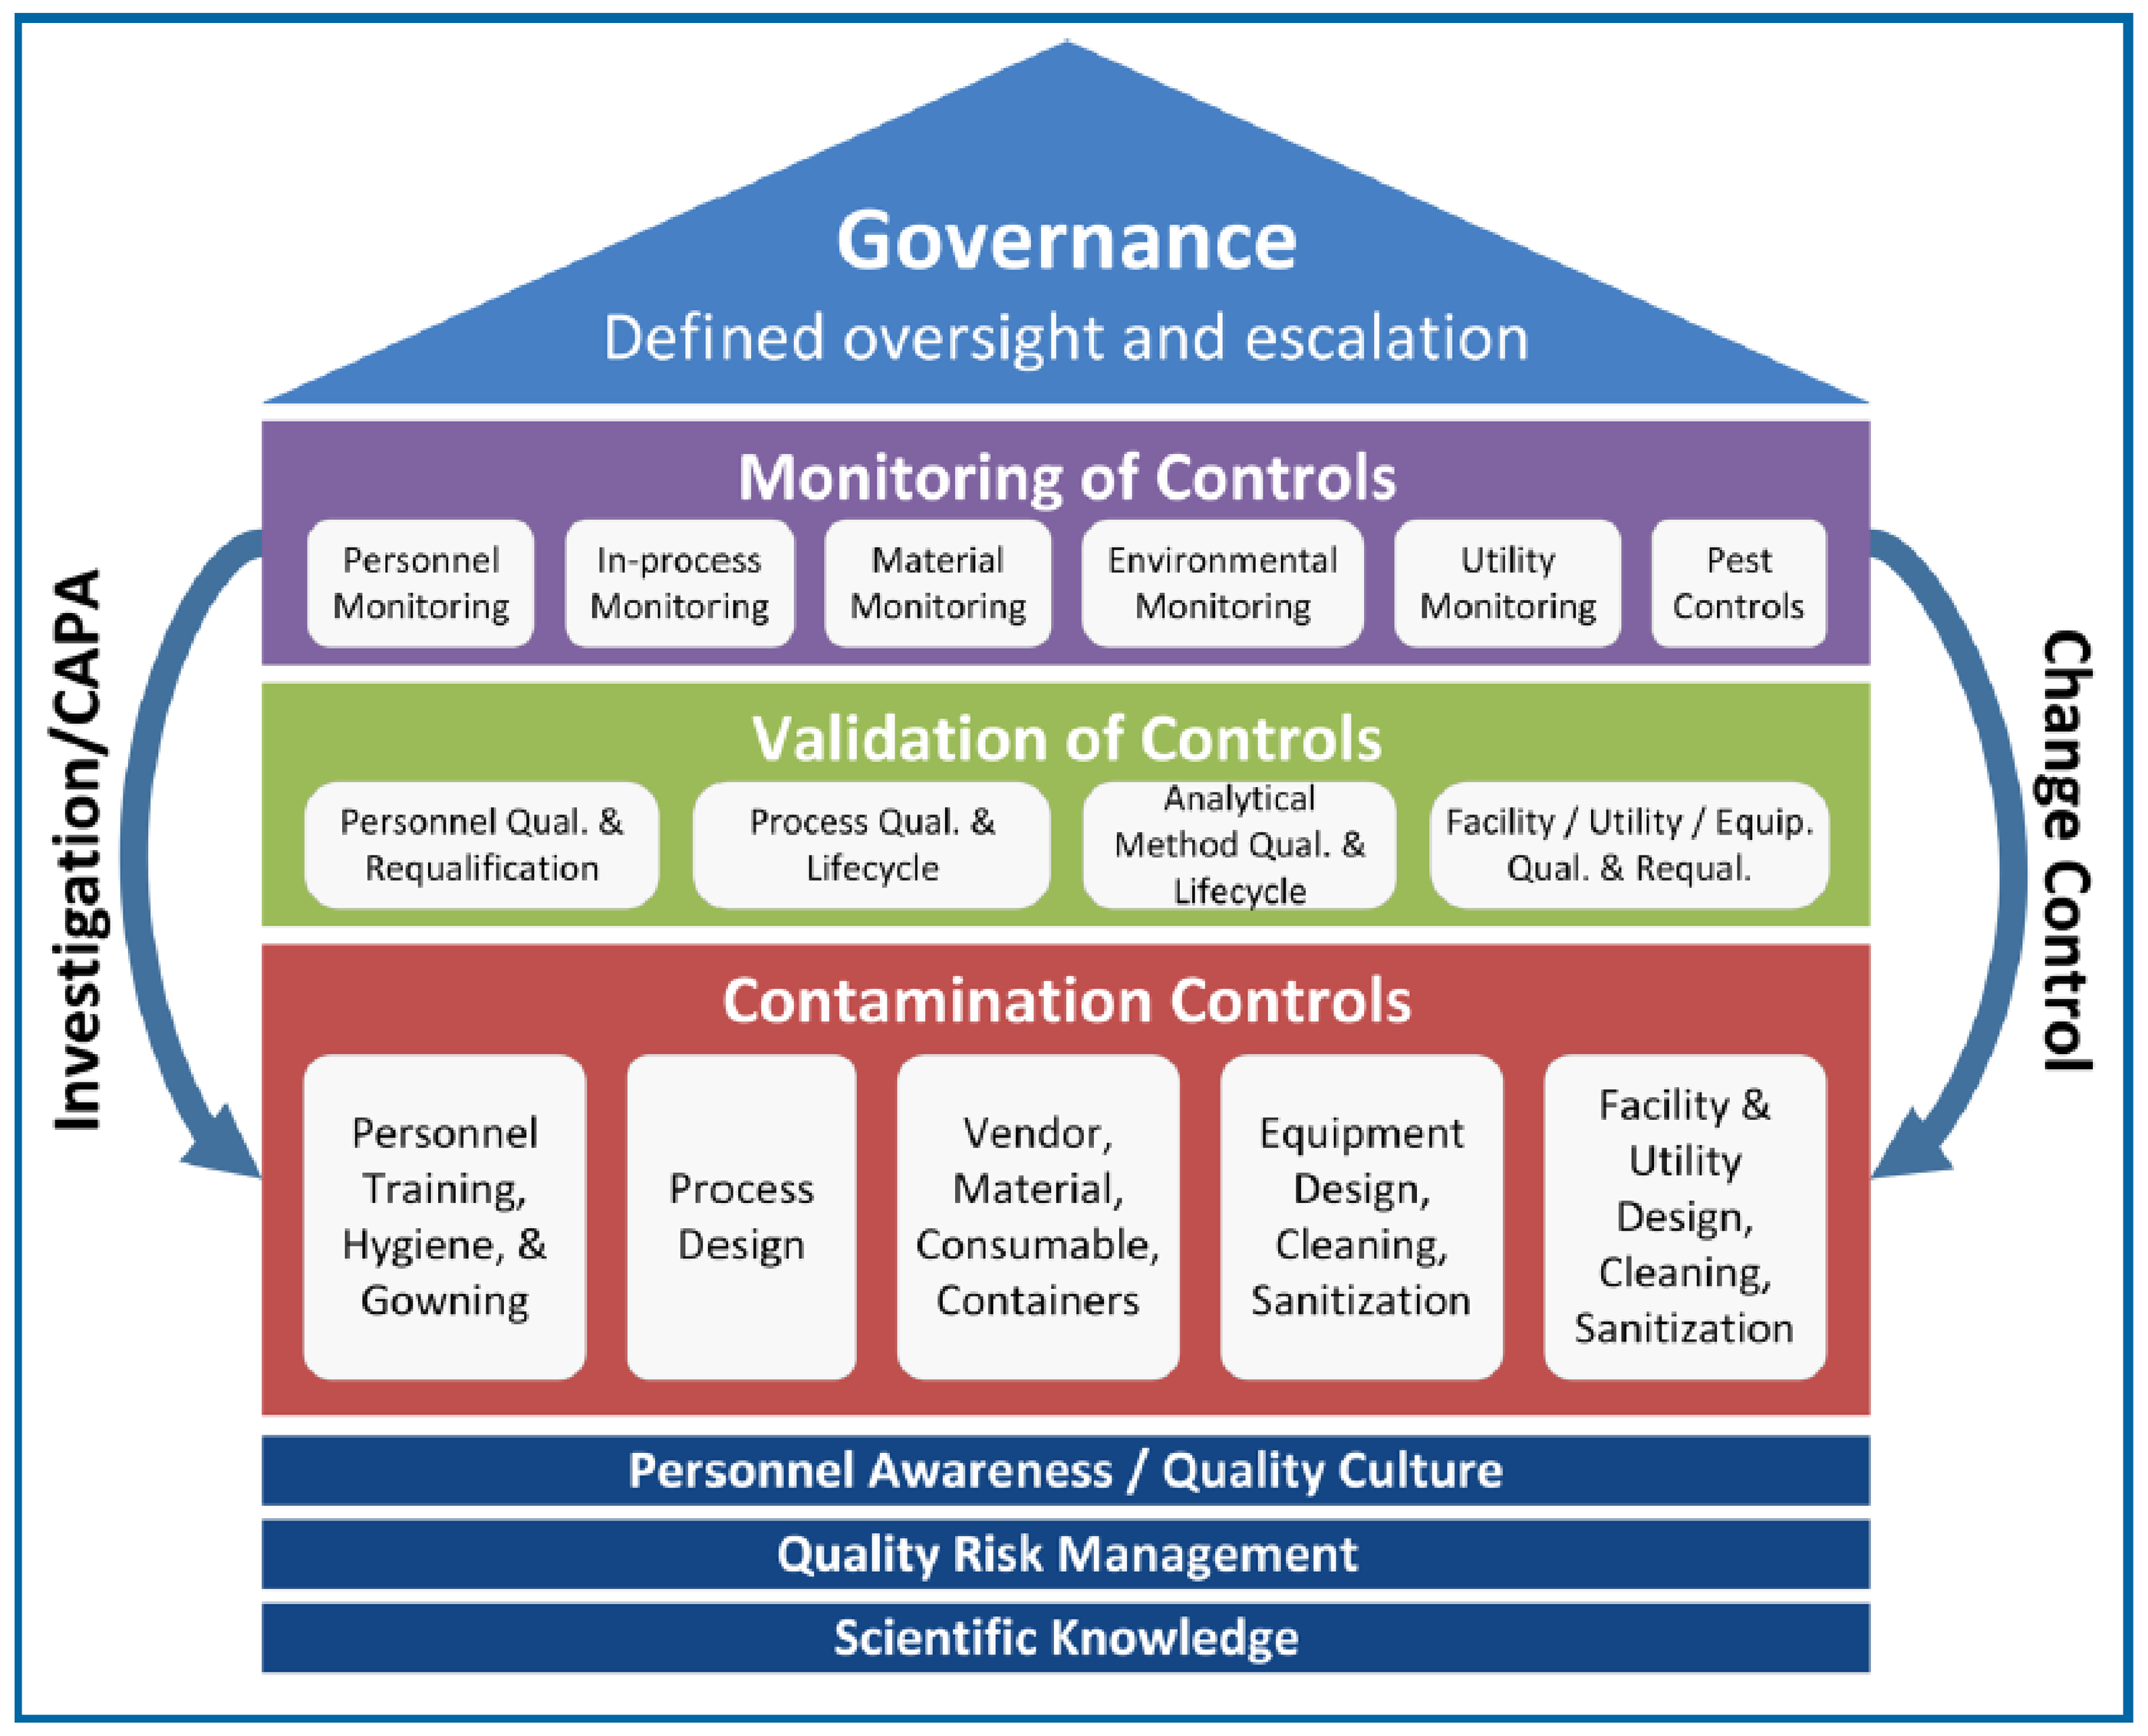
Governance
Defined oversight and escalation
Monitoring of Controls
Personnel Monitoring
In-process Monitoring
Material Monitoring
Environmental Monitoring
Utility Monitoring
Pest Controls
Validation of Controls
Personnel Qual. & Requalification
Process Qual. & Lifecycle
Analytical Method Qual. & Lifecycle
Facility / Utility / Equip. Qual. & Requal.
Contamination Controls
Personnel Training, Hygiene, & Gowning
Process Design
Vendor, Material, Consumable, Containers
Equipment Design, Cleaning, Sanitization
Facility & Utility Design, Cleaning, Sanitization
Personnel Awareness / Quality Culture
Quality Risk Management
Scientific Knowledge
Investigation/CAPA
Change Control

Figure 3.0-1 Elements of a Contamination Control Strategy (courtesy of Sanofi)

"No single failure in one element will result in contamination"
" Design process with redundant controls"[2]

Case Study

Company A is building a new Drug Product facility. As part of contamination control, a CCS is required to be implemented. Below are steps that the company followed to develop a comprehensive and robust CCS.

1. Develop an overall process flow and a detailed process steps for each unit operation within the facility
2. Perform a holistic QRA and identify risks associated with microbial, non-viable particulate, extraneous matter, product-product, and endotoxin/pyrogenic contamination for each process step within the facility
3. Outline existing and proposed controls to mitigate risks
 1. Group controls into pillars of the CCS
4. Develop Action plans for any high and medium risks
5. Develop the CCS document with the preventive and monitoring controls as identified in the QRA.

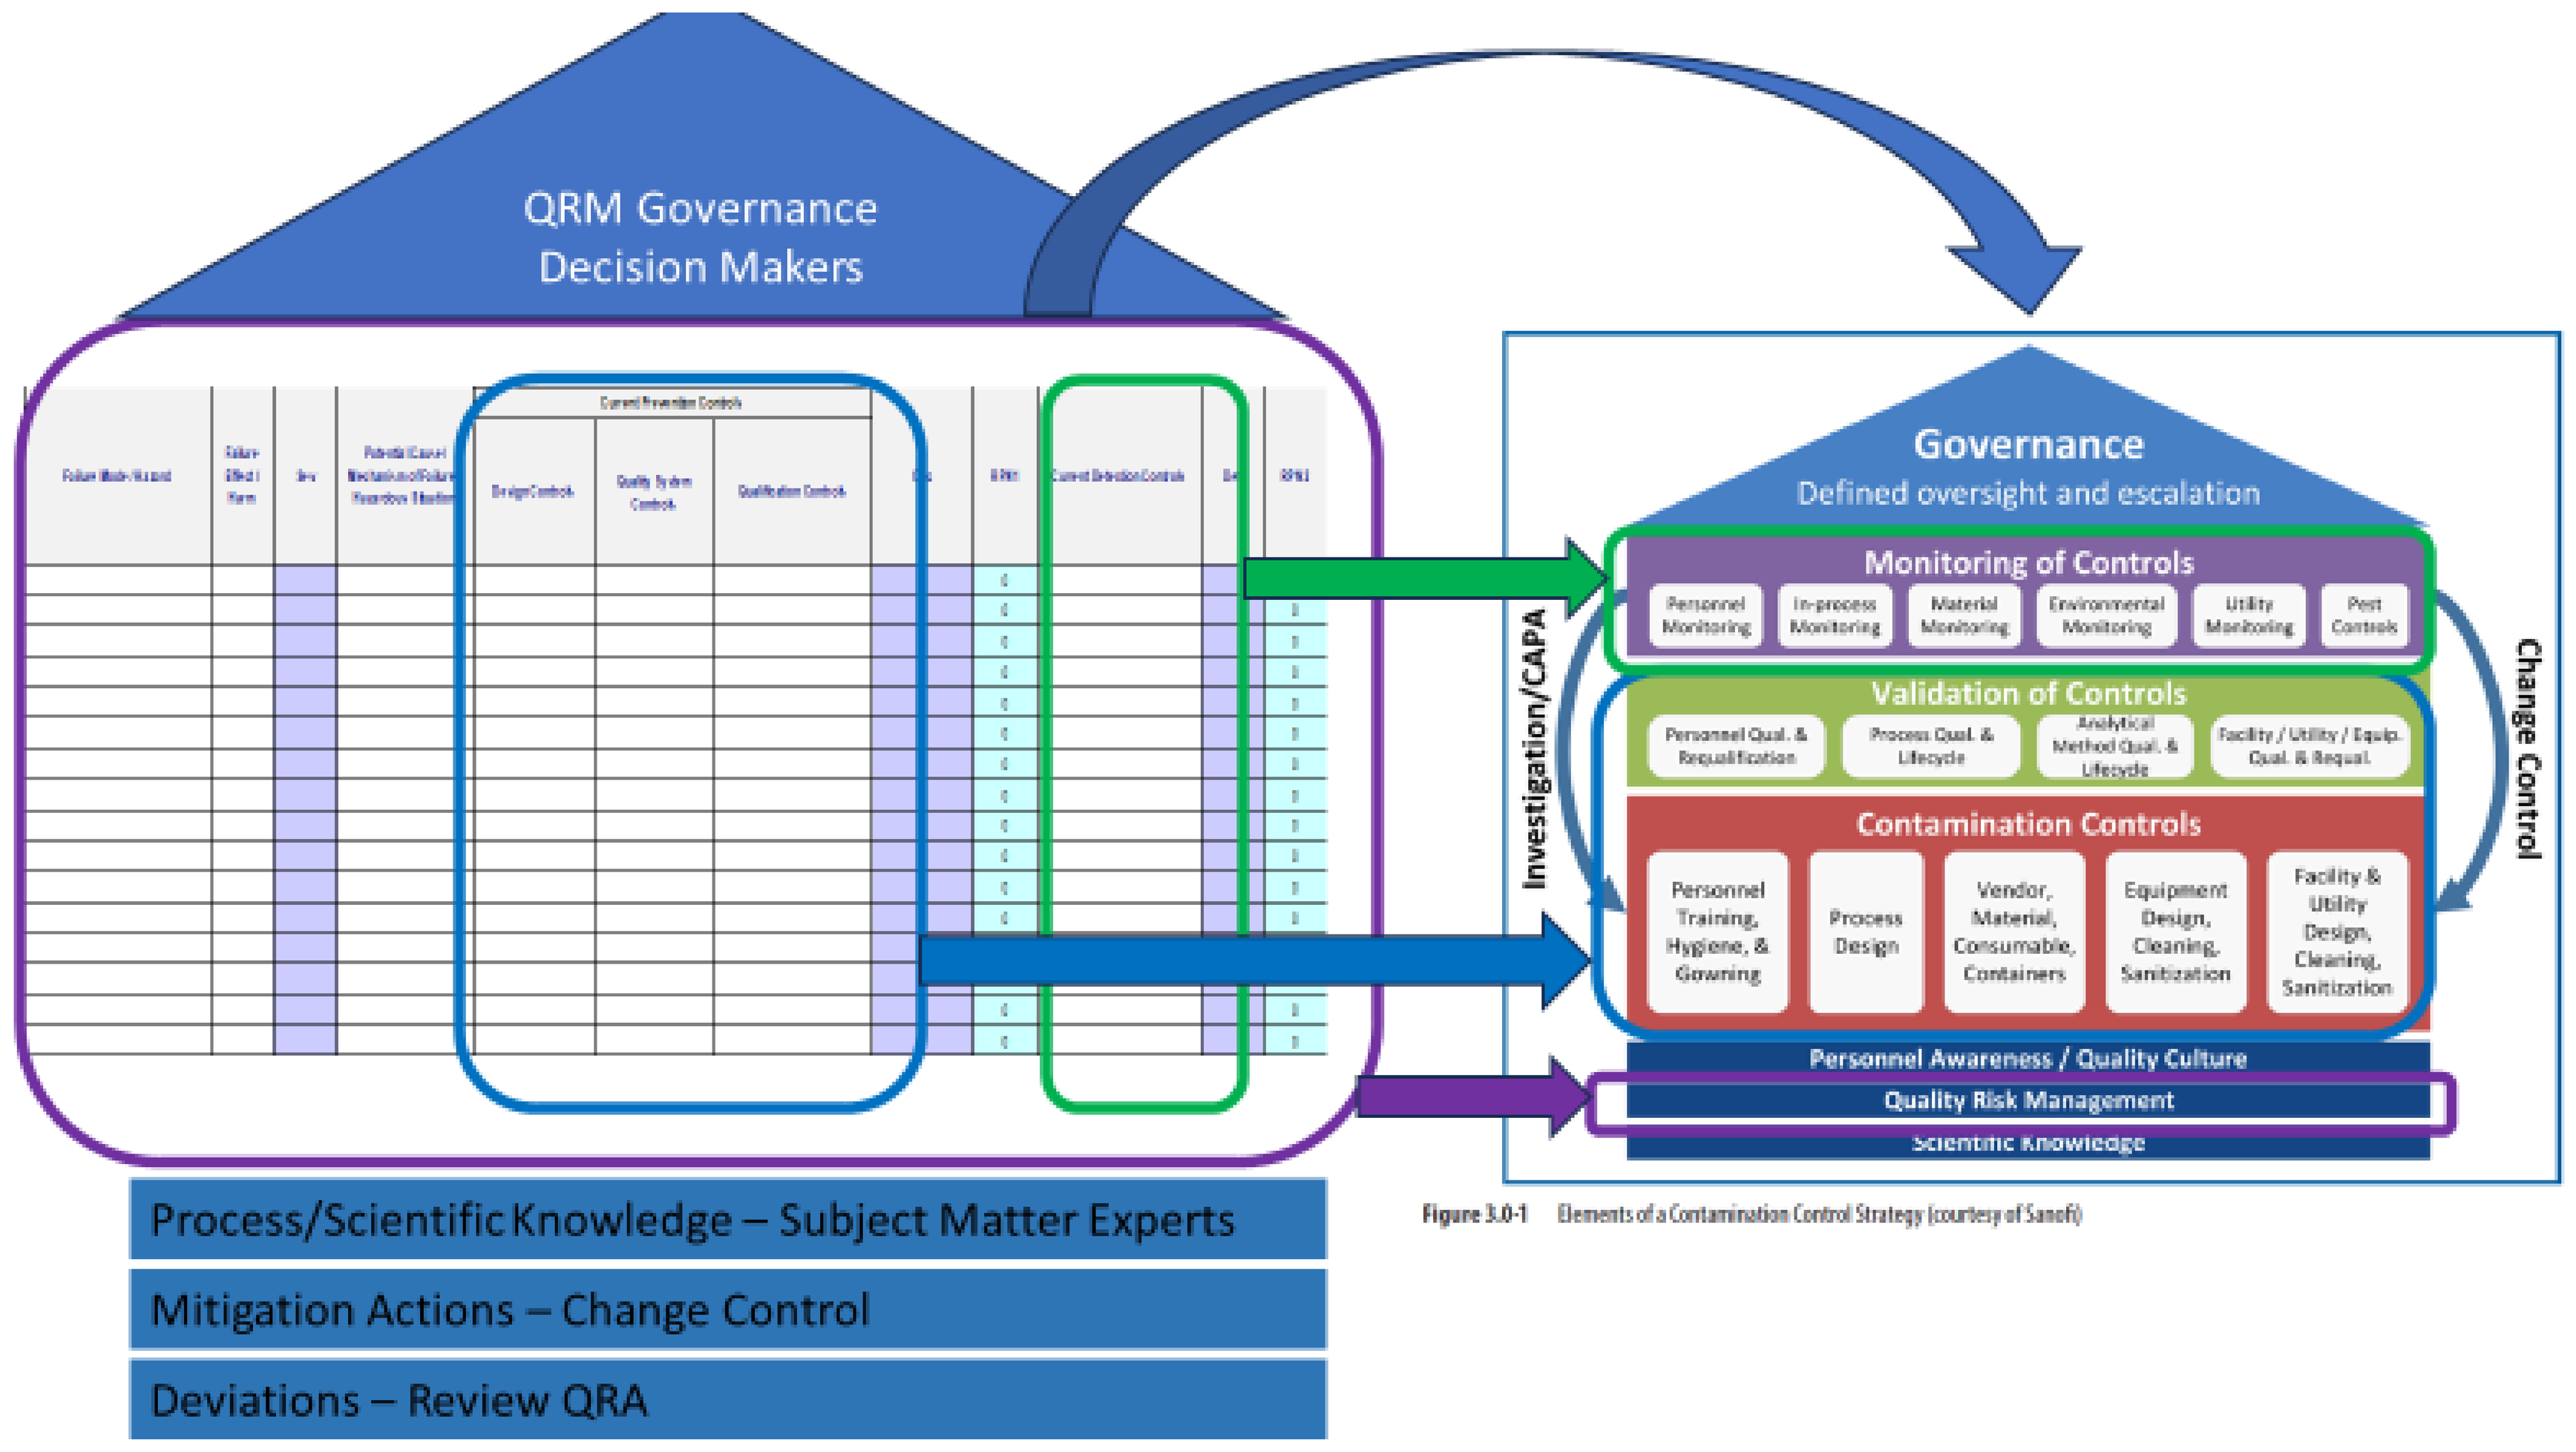
QRM Governance
Decision Makers
Process/Scientific Knowledge – Subject Matter Experts
Mitigation Actions – Change Control
Deviations – Review QRA
Governance
Defined oversight and escalation
Monitoring of Controls
Validation of Controls
Contamination Controls
Personnel Awareness / Quality Culture
Quality Risk Management
Scientific Knowledge
Investigation/CAPA
Change Control
Figure 3.0-1 Elements of a Contamination Control Strategy (courtesy of Sanofi)

Key Takeaways

- QRM principles are the foundation of building a robust CCS
- Key to a robust CCS is to first understand potential risks to your manufacturing process and then establish controls which will translate into the CCS elements.
- A comprehensive CCS plays a pivotal role in managing contamination risks and associated governance.
- A cross-functional team of subject matter experts who have the proficiency in the manufacturing process is essential.

Acknowledgements

[1]Eudralex Volume 4 Annex 1: Manufacture of Sterile Medicinal Products
[2]PDA Technical Report 90 – Contamination Control Strategy Development in Pharmaceutical Manufacturing
[3]ICH Q9 (R1) Quality Risk Management
Manshi Patel, Associate Director, Microbial Control, Merck & Co., Inc., Rahway, NJ, USA

Proprietary