Meaningful & Measurable Risk Assessment Tools for Environmental Monitoring Site Selection Program

Vanessa Vasadi Figueroa, MA,Chief Microbiologist, VVF Science®

INTRODUCTION

The role of Environmental Monitoring has evolved alongside the manufacturing processes and filling technologies its aims to monitor, and so should the risk assessment tools we implement for establishing this important program. Sample site selection, appropriateness of sampling methods, sampling volumes and sampling frequencies are all important components of contamination control for a facility and must be evaluated as appropriate using a robust risk assessment. The types of environmental monitoring required for a robust program will vary based on the type of operation, frequency in which that operation is performed, and the level of risk associated to the process. Developing a meaningful risk assessment tool and systematically applying it to measurable risk rankings for six applicable categories in biopharmaceutical manufacturing will be critical to the outcome of the Environmental Monitoring Risk Assessment (EMRA). The process for scoring each of the six categories, systematic evaluation of contamination probability and example outcomes are provided to demonstrate how EM sites could be mapped throughout an ISO 5 and 7 cleanroom areas, thus ensuring adequate criteria and fair assessment are applied in each case. The methodology for this risk assessment tool can be adapted for various stages, including initial site qualification, evaluating Environmental Monitoring Performance Qualification (EMPQ) results, or periodically updating monitoring requirements during routine operations. Whichever methodology is defined in the protocol should be incorporated into the local site procedure, intended to be executed on a routine basis, or as needed due to changes to the facility, equipment, process, or EM trends observed. It is recommended that site selection be considered during assessment of applicable change controls throughout the year. A periodic review of the EM risk assessment should also be performed as part of routine environmental monitoring trend reports to determine if the sampling frequencies and locations remain appropriate for the detection of potential environmental contamination.

HIGH LEVEL PROCESS OVERVIEW

PREPARATION AND PLANNING

Assemble cross functional team of experts, describe roles & set realistic milestones.

Assemble relevant documents: classifications, MPP flow maps, gowning & occupancy counts.

Identify rooms in scope, assign high-level room description of the activities performed.

Divide each of the rooms into grids with unique identifiers, tally up the grids and layout the grids against the approved facility map.

EXECUTION PART I: DESCRIPTIONS

Distribute the workload by room classification, manufacturing process and grids.

Identify relevant validation documents that will provide key inputs on airflow visualization, aseptic process simulation and facility cleaning.

Each grid gets a detailed process description, relative to the 6 risk categories, which will be used to assign a score to the grid.

Score each of the grids against risk ratings, considering the room classification.

EXECUTION PART II: RISK SCORING

1.Production processes, process flow & types of activity,

2.Proximity to open product or product contacting materials,

3.Aseptic interventions or manipulations required by personnel or their operations,

4.Number of personnel & their traffic patterns, as well as material & equipment flow.

5.Room, equipment & surface accessibility to cleaning/disinfection.

6.Historical EM results from two-year period, including CCR.

TECHNICAL EVALUATION

Accumulate the overall risk score to establish overall rank, and this will be overlaid to a heat map for visualization.

Review the visual heat map against the scores, considering the room classification to ensure all risk rankings were applied consistently. Adjust where needed.

Once the grid scores were established, these are used to determine suitability for the EM locations throughout the facility.

Compare to High risk, Medium risk and Low risk grid guidance. Very Low risk may apply.

ASSIGNING RISK

High-risk grids must have in place adequate representation for all sampling methods – viable surface, viable air, and total particulate.

Medium-risk grids should have in place adequate representation for all sampling methods – viable surface, viable air and total particulate either directly inside the grid or immediately adjacent.

Low & Very Low risk grids may or may not warrant sampling and should be evaluated against grids nearby.

RISK EVALUATION TABLE: 6 CATEGORIES

RISK RATING	TYPE & LEVEL OF PRODUCTION ACTIVITY	PROXIMITY TO OPEN PRODUCT / PROCESS	INTERVENTIONS & INTERACTIONS BY PERSONNEL/ OPERATIONS	FLOW OF PERSONNEL, MATERIAL OR EQUIPMENT	ROOM, EQUIPMENT & SURFACE CLEANABILITY	2Y HISTORICAL EM DATA & CONTAMINATION RECOVERY RATE
1 – Low (rating of 1 for all categories)	No process or low production activities; less than 2 hour duration.	Closed processes, with no product or process exposure to environment. Proximity greater than 60cm (24 in).	No or few personnel interactions or aseptic operations.	Infrequent flow of personnel, material, or equipment. Room occupancy with 0-2 persons	Open area, no accessibility issues for disinfection. Equipment present are easy to clean (i.e., SS table).	3 Out-of-Alert-Level (OOL) results in a two-year time period
2 – Medium (rating of 4 for the proximity to open product factor)	Production and processing occur less frequently but with activity; 2-6 hour duration.	Open processes w/ short duration (< 5min) & down stream controls. Proximity b/w 30cm (12 in) & 60cm (24 in).	Personnel interaction occurs but not frequent; minimal aseptic operations.	Personnel, material and/or equipment move through the area; infrequent. Room occupancy with 3-7 persons	Small or crowded room that is not easily accessible for disinfection. Equipment present; not easy to clean	More than 3 but less than 9 Out-of-Alert-Level results in two-year time period
4 – High (rating of 8 for the proximity to open product factor)	High production area with increased activity; more than 6 hours duration.	Fully open process aseptic operation & controls, filling, critical change parts or components. Proximity less than 30cm (12 in).	Frequent personnel interaction and frequent aseptic interventions in the critical space.	Personnel, material and/or equipment move through the area; frequently. Room occupancy > than 7 persons.	Difficult to clean room or equipment due to layout, process complexity or requires localized clean due to small parts.	More than 9 Out-of-Alert-Level results in two-year period. If critical (Grade A) are monitored, any recovery is OOL.

DISCUSSION

A comprehensive risk assessment should serve as the foundation for modifying or establishing an Environmental Monitoring (EM) program. When introducing new sampling locations or modifying existing sites, each classified area must be evaluated to ensure compliance with minimum regulatory requirements before any reductions or additions are made. Different risk levels correspond to different sampling rates. In general, high-risk areas should be sampled each production shift for Grade A, medium risk areas should be sampled per process for Grade B areas and weekly for Grade C areas, low-risk areas should be sampled weekly, and very low-risk areas should be sampled monthly. Regulatory guidance, such as the EU GMP Annex 1, outlines clear expectations for maintaining contamination control, and these should guide the program's evolution. Only after confirming compliance should changes be considered, ensuring the program remains dynamic and adaptable to the specific risks of each area. The selection of sampling methods—whether viable air sampling, surface sampling, or total particulate monitoring—should be carefully considered. Ensuring that the methods chosen are validated for detecting microbial contamination is essential. For viable air sampling, equipment qualification is particularly important, as it confirms the recovery capabilities of the chosen methodology. Proper validation not only ensures the detection sensitivity of air samplers but the overall integrity of the EM program. For surface sampling, innovative tools such as Enverify™ can provide valuable data to support recovery validation, ensuring that contamination is adequately captured from critical surfaces. Whenever changes are made to EM site selection, detailed justifications must accompany the decision. This includes clear reasoning for adding new sites, often driven by risk-based considerations, as well as transparent explanations when sites are removed.When reducing sites, it is critical to demonstrate how the surrounding sampling locations will continue to capture any potential contamination, ensuring that the overall contamination risk remains under control. Providing this transparency not only strengthens the rationale for changes but also supports future compliance audits by regulatory bodies, offering documented evidence of why certain decisions were made. Furthermore, capturing a historical perspective on changes to the facility, processes, or contamination control techniques is considered a best practice. This historical tracking provides context for understanding how and why the EM program has evolved over time, which can be invaluable during regulatory inspections. Historical documentation also helps link changes in the manufacturing environment—such as new equipment, layout modifications, or updated techniques—to the corresponding adjustments in the EM program, ensuring a proactive rather than reactive approach to contamination control. Systematic risk-based evaluations must drive modifications to the EM program, supported by validated sampling methodologies and transparent justifications for all changes. This approach not only ensures the ongoing robustness of the EM program but also enhances the ability to maintain regulatory compliance and safeguard product quality.

RECOMMENDED DOCUMENT FLOW

#	Doc Type	Description	Program
01	Protocol	EM Risk Assessment and Site Selection Protocol	W
02	Attachment	List of Rooms and Grid Pattern distribution in Facility	X
03	Attachment	Room, Grid and Process Descriptions with Detailed Risk Scoring	X
04	Attachment	Facility Map Layout with Numbered Grid Distributions	A CAD
05	Attachment	Summary of Risk Ratings, & Graphical Representation of Results	X
06	Attachment	Summary of Added, Reduced, Modified Sampling locations	X
07	Attachment	Facility Heat Map Visualization of Final Grid Risk Scoring	A CAD
08	Final Report	EM Risk Assessment & Site Selection Final Report	W

SEQUENCED PROCESS RESULTS

Room Grid	Class	Process Activity Description performed in the Room	Risk Score Justification Details	Risk Category 1: Level of Production Activity	Risk Category 2: Proximity to Open Product	Risk Category 3: Interventions by Personnel/ Operations	Risk Category 4: Flow of Personnel, Material or Equipment	Risk Category 5: Room, Equipment & Surface Cleanability	Risk Category 6: Historical EM data (Two Years)	Final Risk Score: Category 1x2x3x4x5x6	Rating Score: very low-risk = 1 thru 4, low-risk = 8 thru 32, medium-risk = 64 thru 512, high-risk = 1024 thru 8192
101-G01B	Grade B	Aseptic gowning room where final gown and sterile gloving are performed. Adjacent access to aseptic corridor.	**Production Activity:** Final gowning and gloving, this is also a regloving station where needed for entrance to the aseptic corridor. Cumulative time of production activities between 2-6 hours. **Proximity to Open Product:** None **Interventions by Personnel/ Operations:** Frequent donning of sterile garments and changing final sterile gloves. **Flow of Personnel, Material or Equipment:** Multiple personnel frequently enter/exit grid for gowning practices. Max Personnel 4. **Room, Equipment & Surface Cleanability:** Moderately difficult to clean due to the quantity of gowning supplies that are routinely stocked on racks and gloveboxes that require removal during cleaning. Bench is located in the room. **Historical EM data:** 2 alerts, and 3 actions in the past 2 years, CRR 3.7%.	2	2	4	4	2	2	256	medium risk
102-G01A	Grade A	Grade A Filling environment that is enclosed with oRABS where empty vials enter from depyro. Filler set-up occurs through open doors using aseptic practices. Once filling begins, aseptic interventions occur predominantly through glove ports; rare open door interventions are permitted as needed.	**Production Activity:** Incoming accumulator table where glass is introduced to the aseptic line. Activity duration greater than 6 hours up to 3 times per week. **Proximity to Open Product:** Proximal, within 12 inches of open empty vials intended for sterile liquid. **Interventions by Personnel/ Operations:** EM set-up, EM Interventions. High risk intervention of removing fallen empty vials. **Flow of Personnel, Material or Equipment:** Vials transition, forceps/EM materials enter through RABS gloves, interventions through RABS during filling, change parts removed when not filling. No personnel movement. **Room, Equipment & Surface Cleanability:** Surfaces are SS and glass which are smooth and easy to clean. Multiple small/hard to access spaces on table that are difficult to clean. Following open door intervention, areas contacted, RABS door, and RABS glove are all disinfected. **Historical EM data:** 0 alert and 0 action contact plate, 0 alert and 0 action active air and 0 alert and 0 action particulate over two years. CRR 0%.	4	8	4	4	4	2	4096	high risk

Example process descriptions for a medium risk grid, and a high risk grid in the dummy facility. This illustrates the in-depth process needed to systemically define the grids, describe them and evaluate each grid against the 6 risk categories.

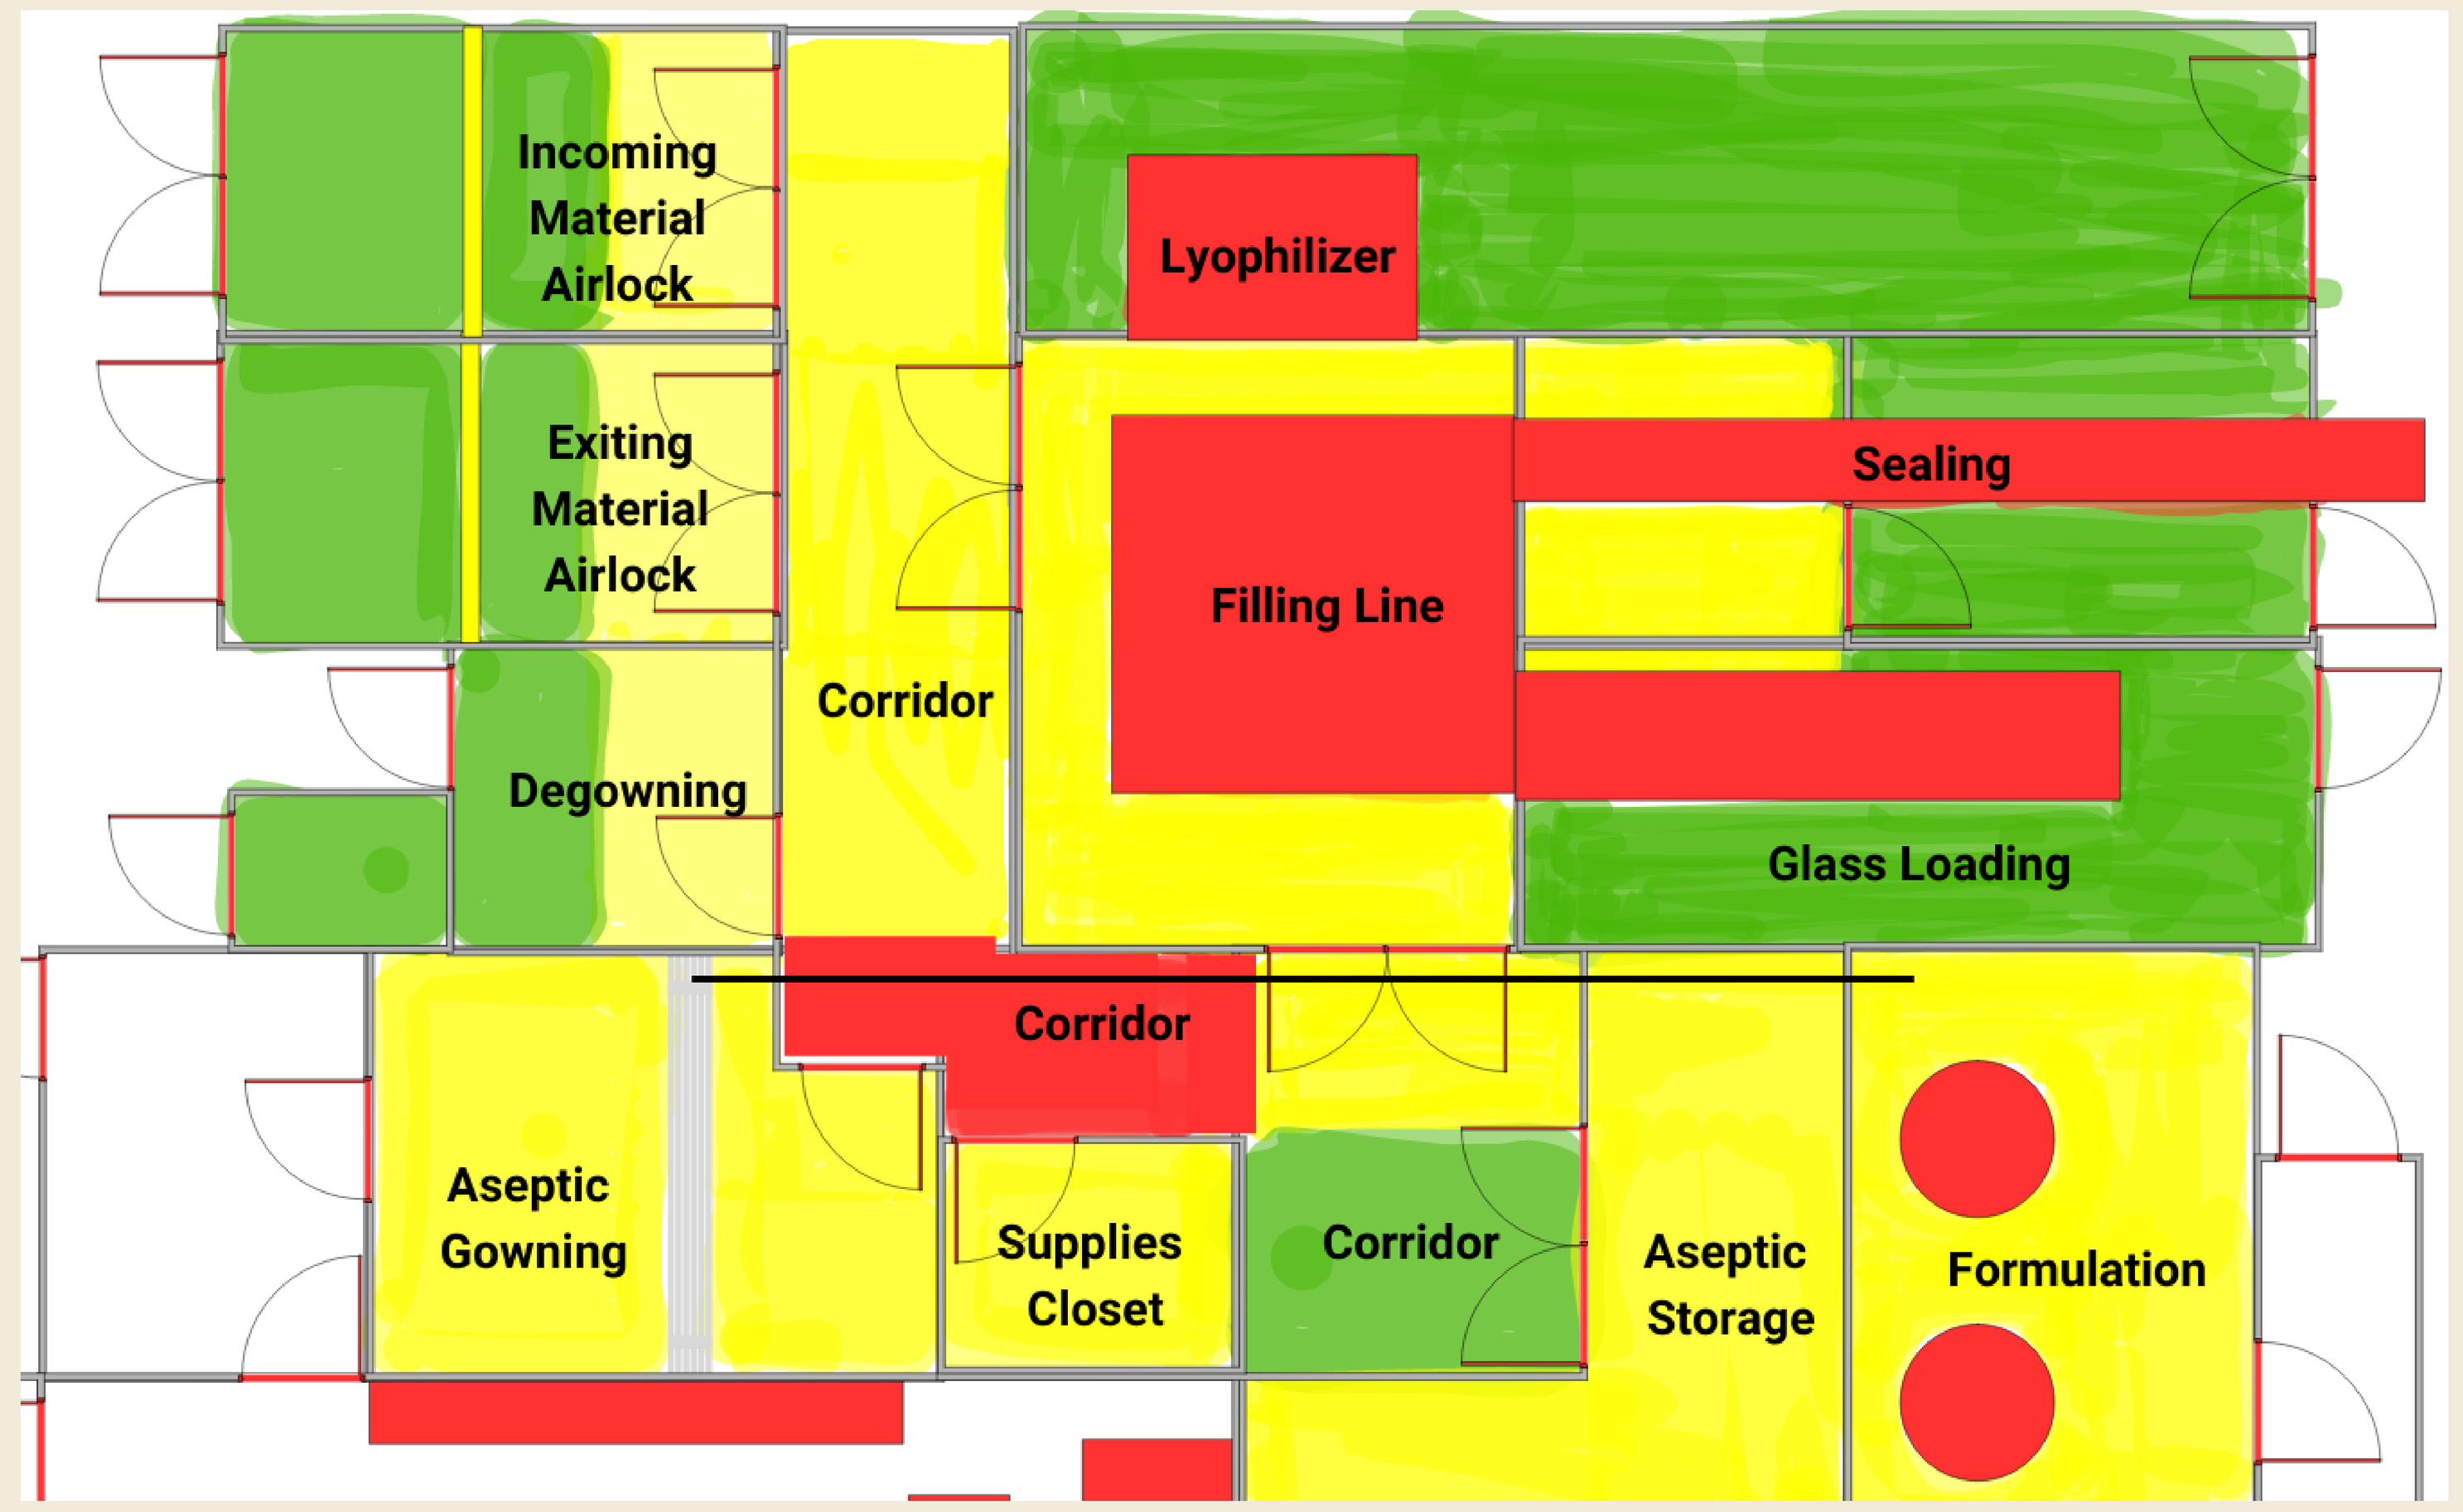

Incoming Material Airlock
Lyophilizer
Exiting Material Airlock
Sealing
Filling Line
Corridor
Degowning
Glass Loading
Corridor
Aseptic Gowning
Supplies Closet
Corridor
Aseptic Storage
Formulation

Aseptic Filling Line, example of the heat map distribution of a dummy facility. This is for illustration purposes only and is not to be replicated. Red is high risk, yellow is medium risk and green are low risk grid profiles. There are no very-low risk grids in this portion of the facility.

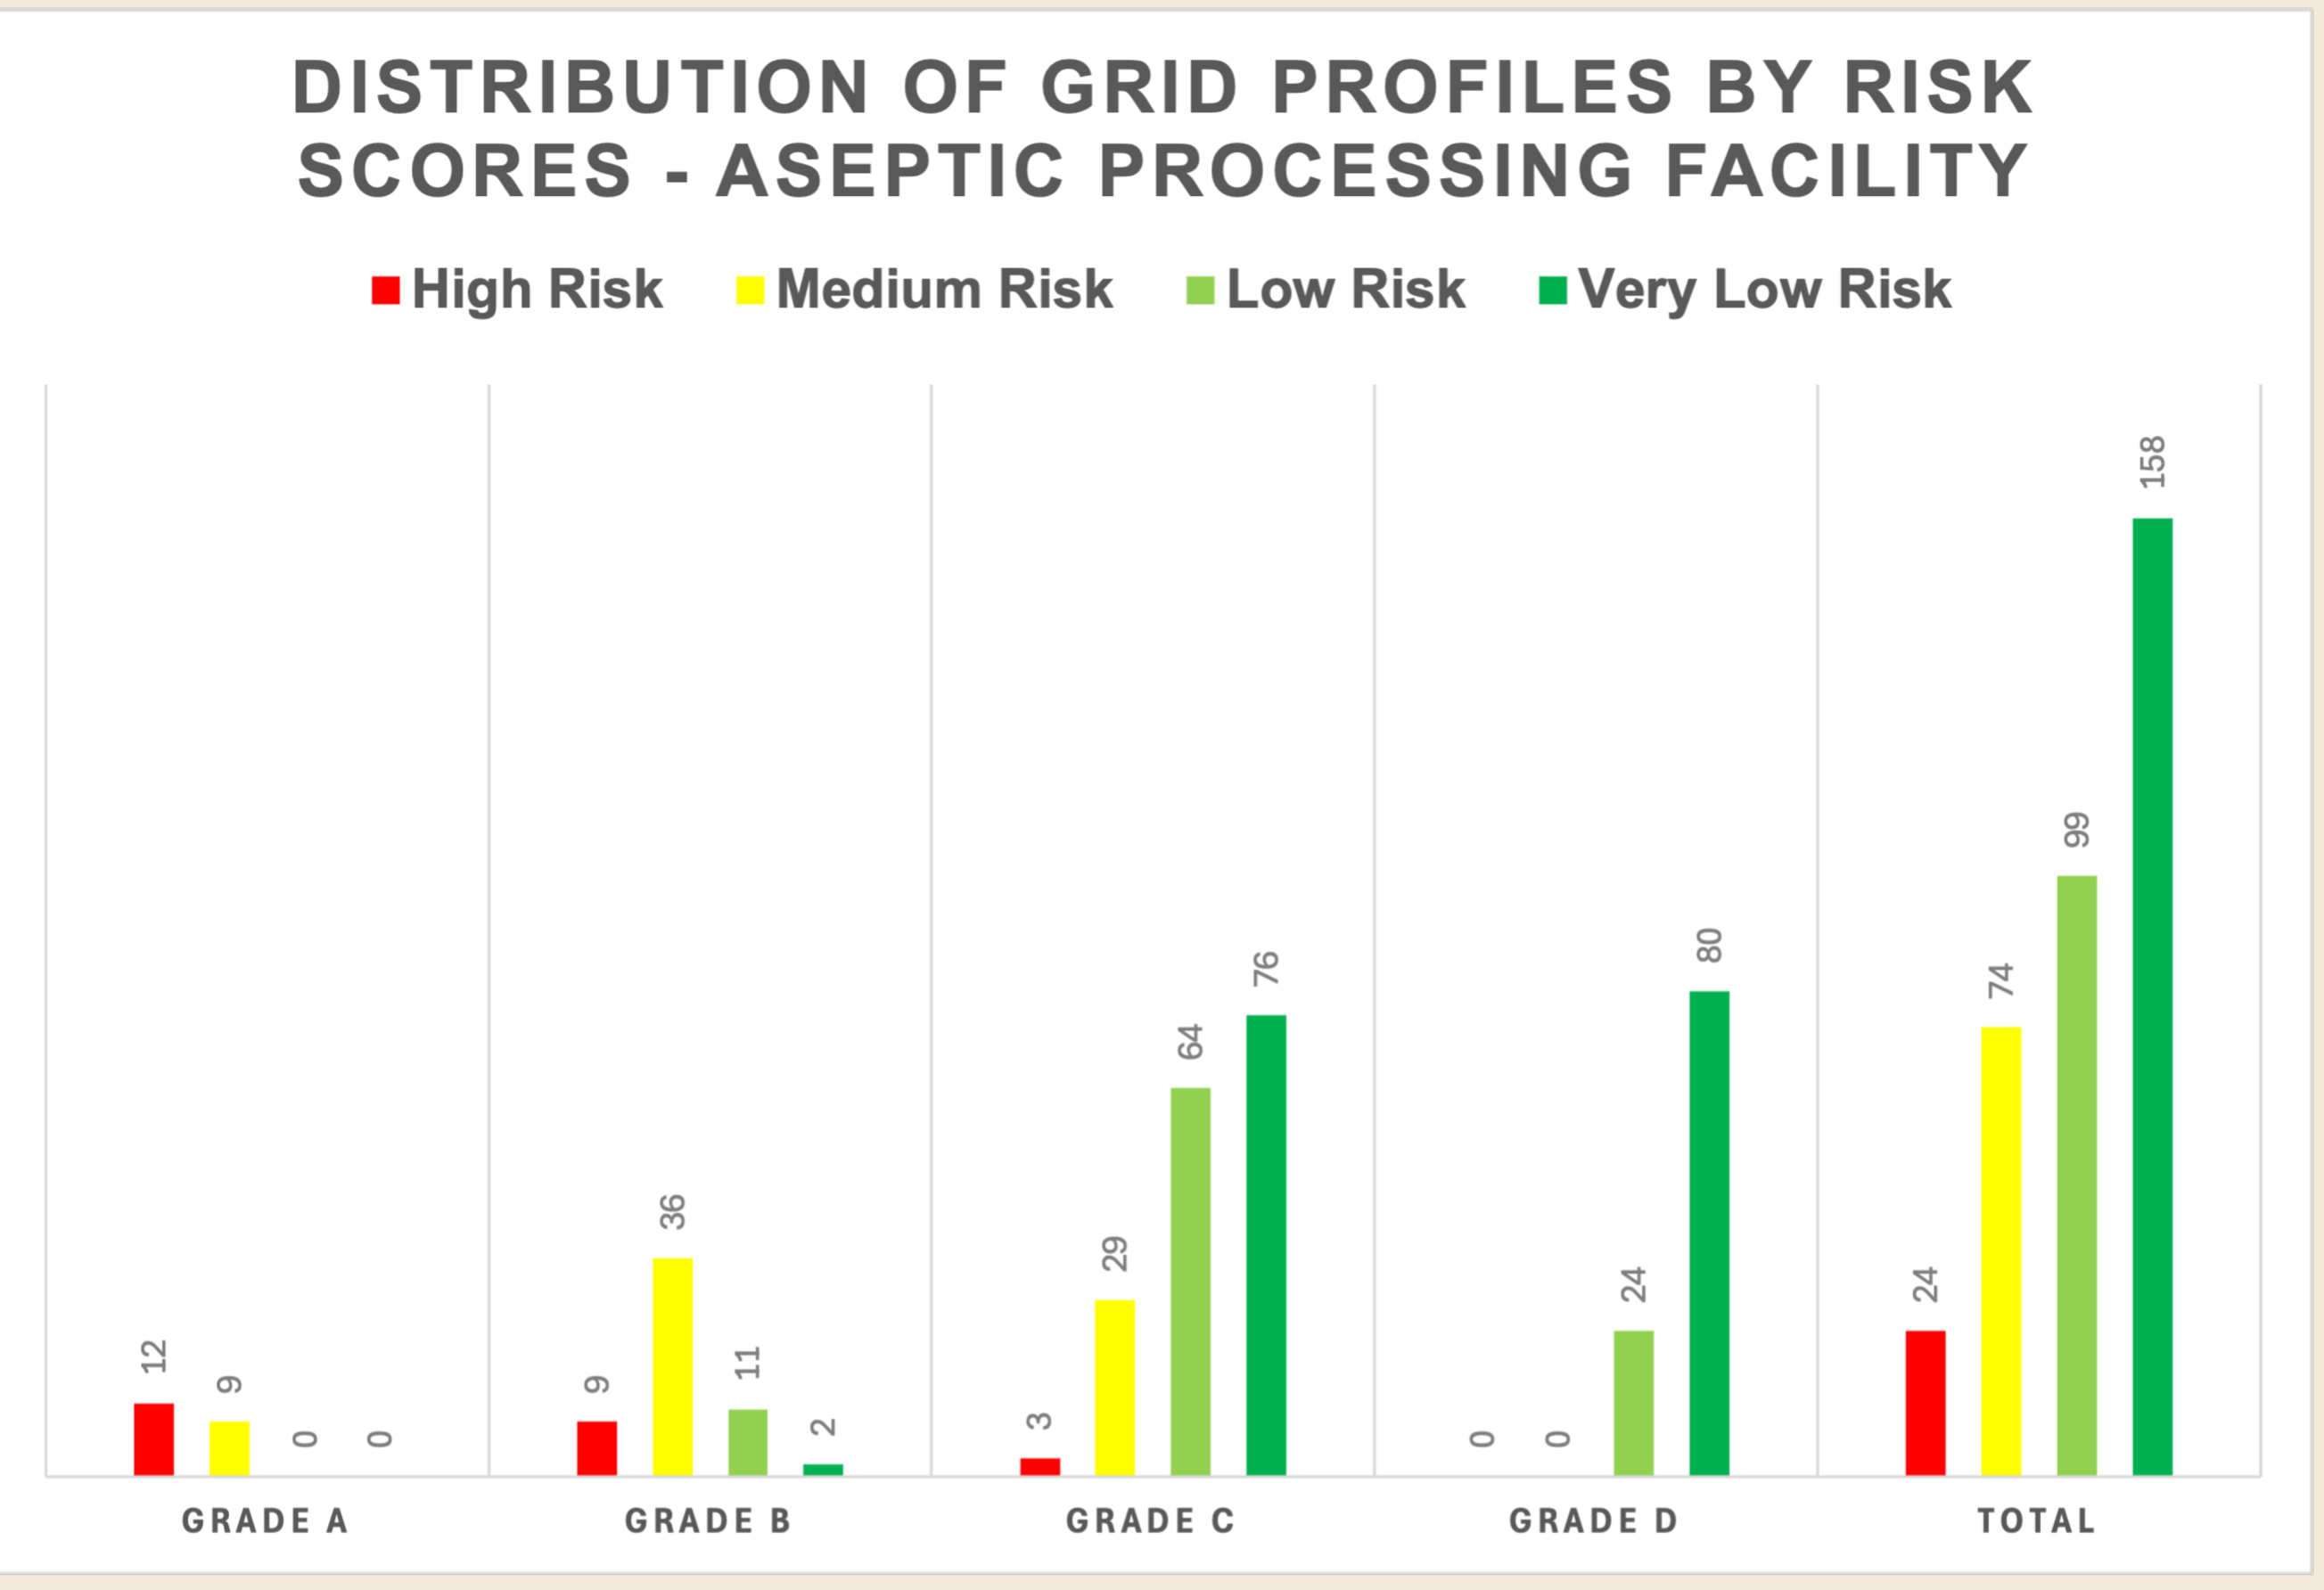

DISTRIBUTION OF GRID PROFILES BY RISK SCORES - ASEPTIC PROCESSING FACILITY
High Risk
Medium Risk
Low Risk
Very Low Risk
12
9
0
0
9
36
11
2
3
29
64
76
0
0
24
80
24
74
99
158
GRADE A
GRADE B
GRADE C
GRADE D
TOTAL

Distribution of the grid profiles by risk scores. This graph illustrates the number of risk profiles for each classified area in the dummy facility, demonstrating that high risk areas are concentrated to the Grade A predominantly, followed by Grade B & some Grade C areas.

REFERENCES

1. Watson, et al. "Environmental Monitoring: A Harmonized Risk-Based Approach to Selecting Monitoring Points and Defining Monitoring Plans". BioPhorum Operations Group Ltd, November 2020.

2."Fundamentals of an Environmental Monitoring Program". Parenteral Drug Association, Technical Report 13, Revised 2022.

3. U.S. Food and Drug Administration. "Guidance for Industry: Sterile Drug Products Produced by Aseptic Processing — Current Good Manufacturing Practice". September 2004, www.fda.gov/media/71026/download.

4. European Commission. "Eudralex – Volume 4 – Good Manufacturing Practice Guidelines, Annex 1 Manufacture of Sterile Medicinal Products". Brussels, Belgium,2022, health.ec.europa.eu/system/files/2022-08/2022_annex1_en_0.pdf.

5. "Cleanrooms and Associated Controlled Environments – Part 1: Classification of Air Cleanliness by Particle Concentration". ISO 14644-1, 2015, www.iso.org/standard/53394.html.

6. U.S. Pharmacopeia. "Bioburden Control of Nonsterile Drug Substances and Products". USP <1115>, United States Pharmacopeial Convention.

7. U.S. Pharmacopeia. "Microbiological Control and Monitoring of Aseptic Processing Environments". USP <1116>, United States Pharmacopeial Convention.

8. Poster created using BioRender, www.biorender.com and creative visualization formats imported from Canva, www.canva.com.

Created with BioRender Poster Builder